Design of an Innovative Model to Improve Anticancer Therapeutic Treatment Based on a Vascularized 3D Organoids Derived from Patients' Lung Tumors for the Purpose of Precision Medicine

Helene Le[1,2*], Joseph Seitlinger[1], Ysia Idoux-Gillet[1], Véronique Lindner[1], Nadia Benkirane-Jessel[1], Cécile Zaupa[2], Michelle Klein[2,] Jean-Marc Balloul[2], Eric Quéméneur[2]
[1] INSERM-UNISTRA, UMR 1260, Regenerative Nanomedicine (RNM), FMTS, 67000 Strasbourg; website: regmed.fr
[2] TRANSGENE, Boulevard Gonthier d'Andernach, Parc d'Innovation, 67400 Illkirch-Graffenstaden, France; website: transgene.fr
*E-mail: hle@transgene.fr

Poster ID: 13

join.skype.com/aNI0AXB16QrR

INTRODUCTION

Lung carcinoma is a major cause of cancer death, especially non-small cell lung cancer (NSCLC) has the worse prognosis, with survival rate over 5 years below 15%[1]. Unfortunately, there is still a lack of efficient treatments for several patients despite the large number of recently approved therapeutic classes, from chemotherapy to immunotherapy. The objective of the present project is to develop a preclinical/translational model that recapitulates the heterogeneity of tumors in order to better select treatment options, and to support the research for novel drugs with improved properties. We here propose an *in vitro* 3D model derived from patient's tumor biopsies, that reconstitutes the tumor vascularization and the microenvironment directly from patients' biopsies. This advanced assay was designed to match the criteria of drug assessment and screening, and in the longer term to promote the access to personalized medicine for patients with NSCLC.

MATERIAL & METHODS

Methods

Human specimens

Biopsies of lung cancer and healthy tissues were obtained by surgery. Patients' informed consents were managed by the CRB (*Centre de Ressources Biologiques des Hôpitaux Universitaires de Strasbourg*). Samples were confirmed as tumoral or normal tissues following anatomopathological analysis.

Tissue preparation and culture of PDOs

Human samples were washed in PBS and enzymatic digestion was performed at 37° C for 1h with intermittent agitation. After incubation, the reaction media containing samples was transferred to a Falcon tube through 70µm cell strainers. The strained cells were centrifuged and the pellet was resuspended in 1mL of DMEM-F12 (Lonza). The cell suspension was then seeded for a final volume per drop of 40uL in AkuraPLUS™ hanging drop system (96 wells). PDOs formed after 5 days of incubation at 37° C.

Histology and imaging

Healthy and tumoral PDOs were fixed in 4% paraformaldehyde (PFA). They were given to anatomical pathologists to perform IHC and standard H&E staining.

IF

Healthy and tumoral PDOs were harvested and placed in molds with optical coherence tomography (OCT) then frozen. The obtained blocks were sectioned, fixed in 4% PFA, permeated and blocked with 0,1% Triton X-100/1% BSA/PBS for 30min. They were incubated with primary antibodies overnight at 4° C. Primary antibodies were detected by secondary antibodies, incubated for 1h at RT. Nuclei were stained with DAPI (1:2000 dilutions, D9542, Sigma) for 5min. Imaging was performed on a Leica DM4000 B microscope.

Drug screening

PDOs cultured in *in sphero* plates over 5 days were harvested in a ULA-plate. PDOs were then tested with 7 concentrations of taxol and DMSO controls for 2 days. After 2 days, the organoids were transferred in a 96 well plate and the medium was changed to 100µL of fresh PDOs media and 100µL Cell Titer-Glo (Promega). The plates were agitated 10mn at RT prior to luminescence reading.

RESULTS & DISCUSSION

Study Objectives

- To valid the patients' derived organoids (PDOs)
- To establish a multicellular organoid
- To perform drug screening and see the correlation between preclinical responses and clinical responses
- To develop a microfluidic device

Results

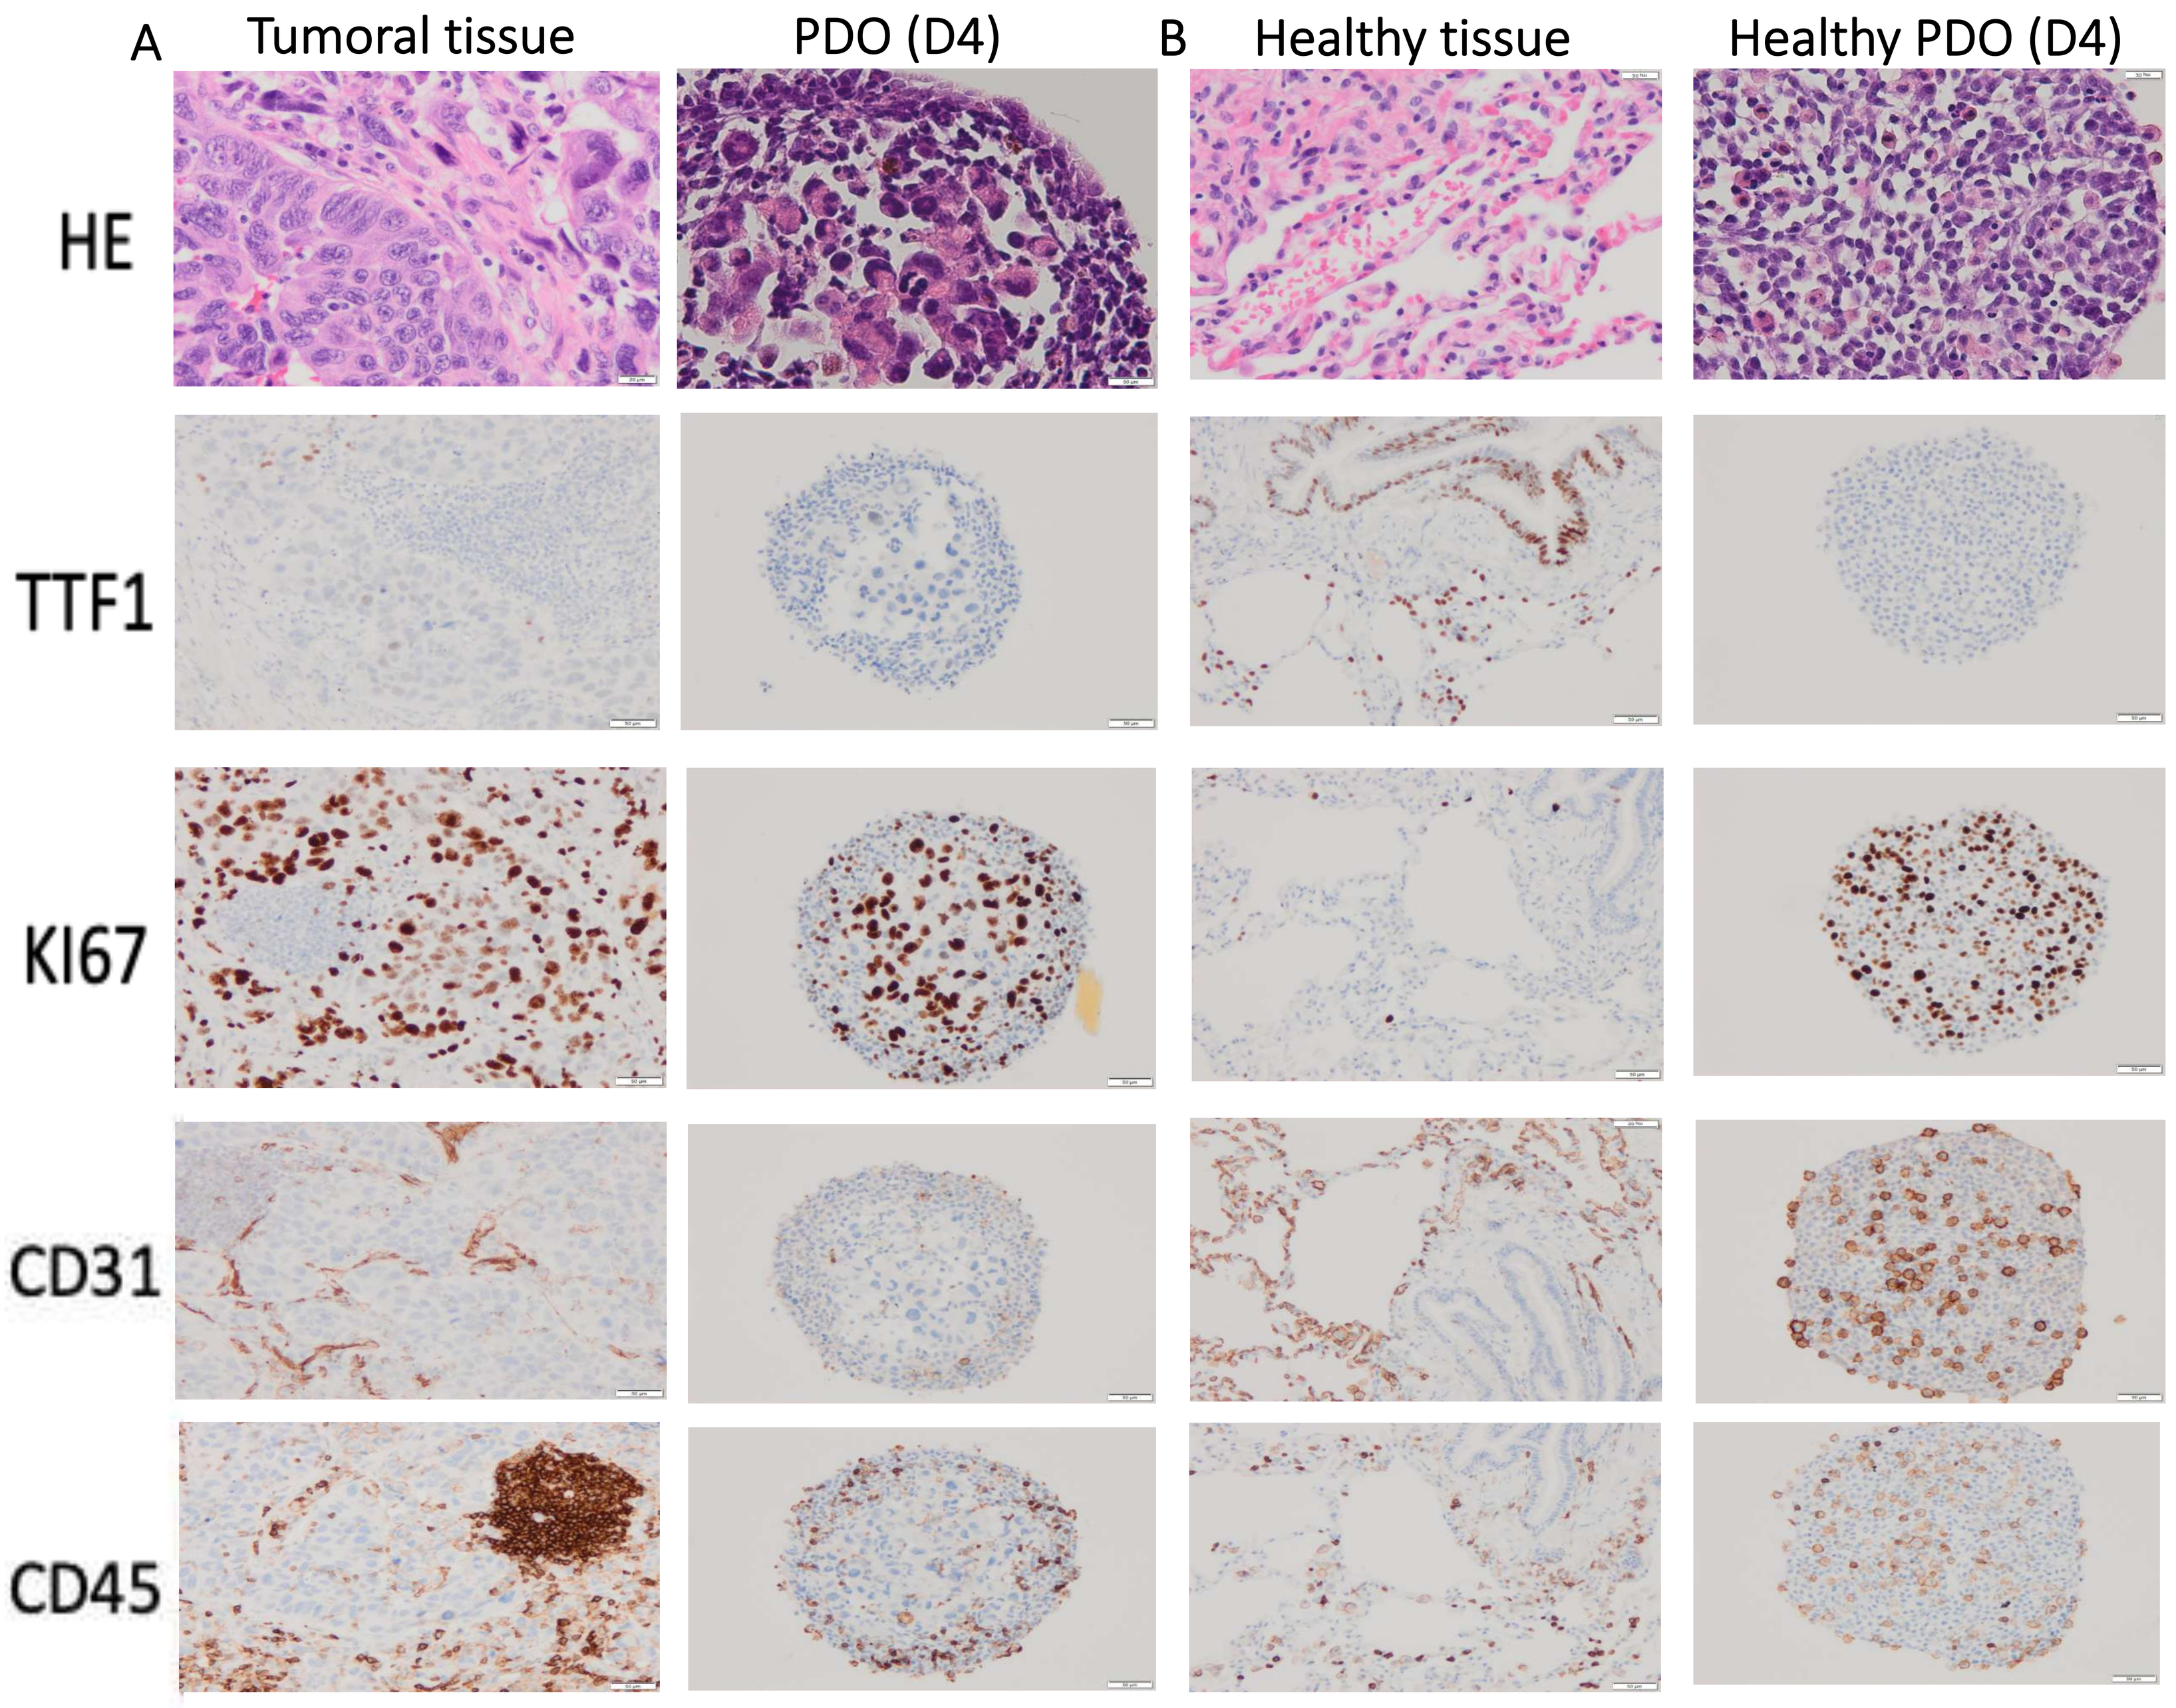
A
Tumoral tissue
PDO (D4)
B
Healthy tissue
Healthy PDO (D4)
HE
TTF1
KI67
CD31
CD45

Figure 1. A, HE-stained & IHC-stained images of a patient's primary tumor and its PDO at day 4. B, HE-stained & IHC-stained images of a patient's original healthy tissue and its healthy PDO at day 4.

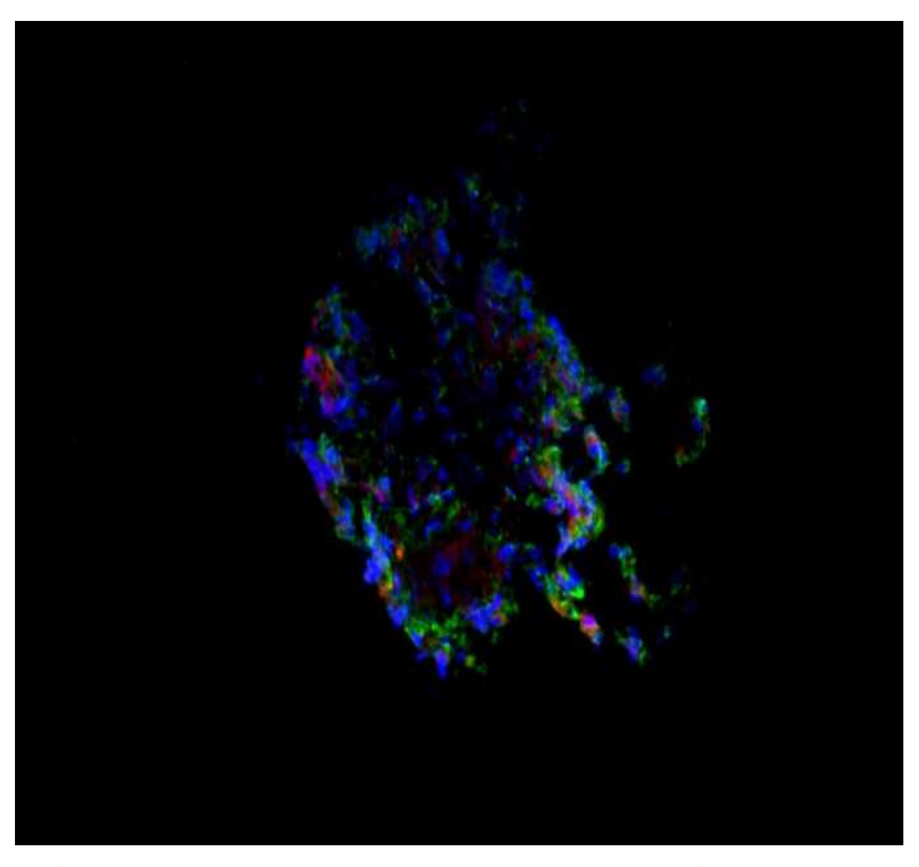

Figure 2. IF images showing the expression of fibronectin (green) and TTF1 (red) of a tumoral PDO. Nuclei (blue) were stained with DAPI

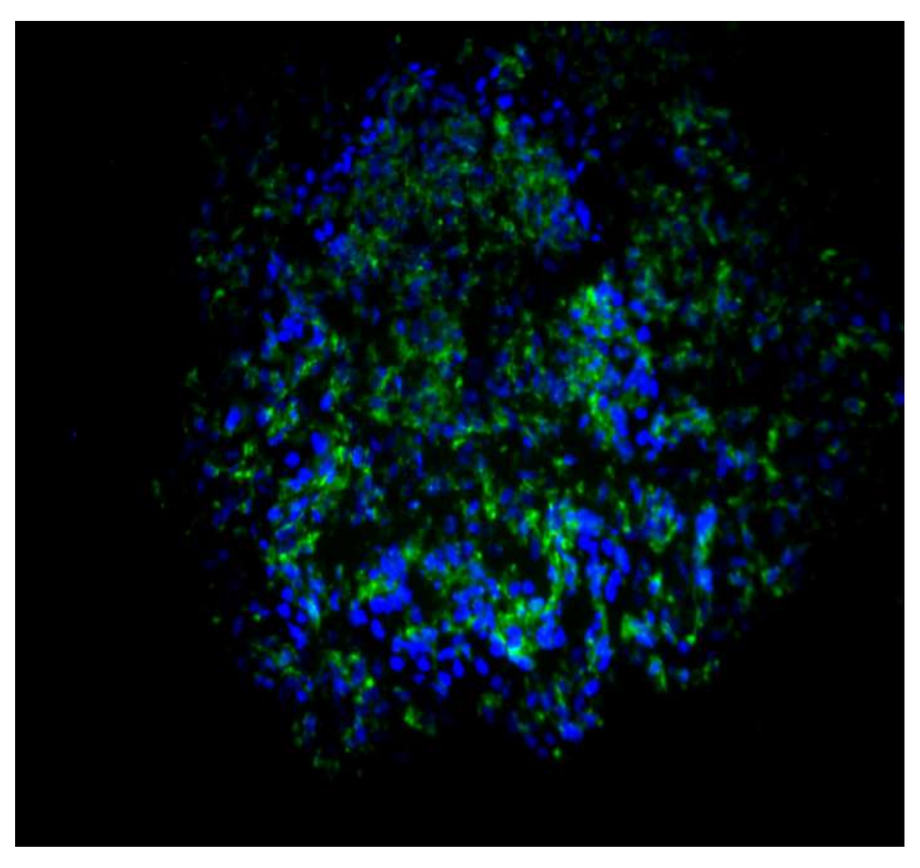

Figure 3. IF images showing the expression of fibronectin (green) and TTF1 (red) of a healthy PDO. Nuclei (blue) were stained with DAPI

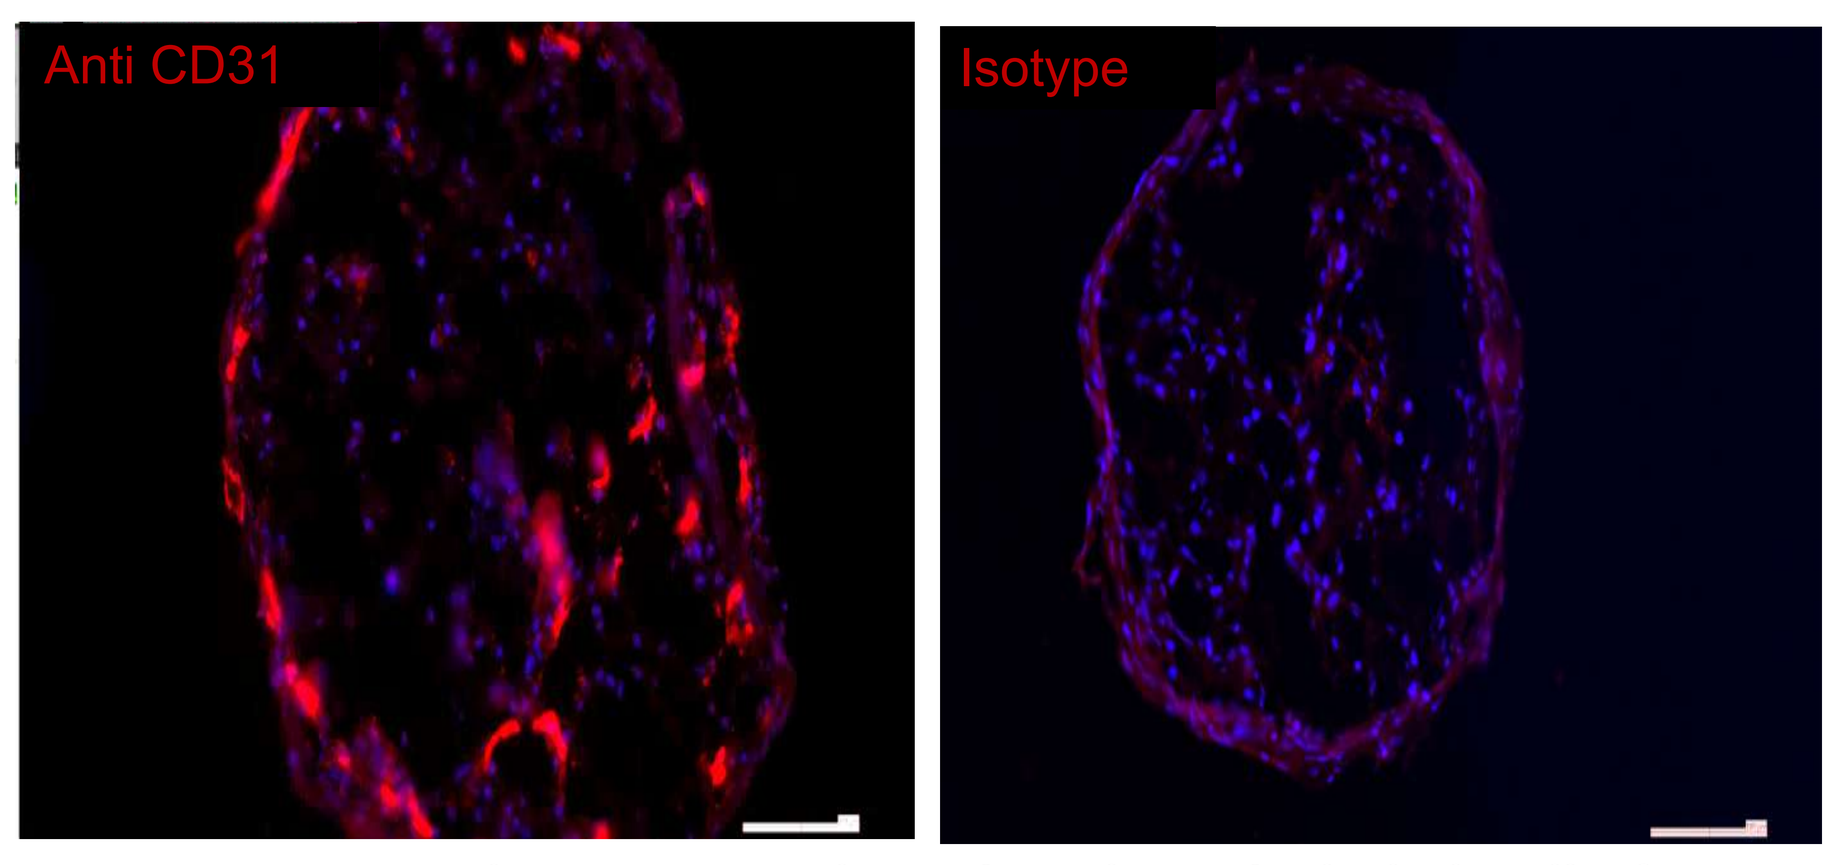
Anti CD31
Isotype

Figure 4. Tumoral PDOs co-cultured with endothelial cells for 29 days. IF images showing the expression of CD31 (red). Nuclei (blue) were stained with DAPI

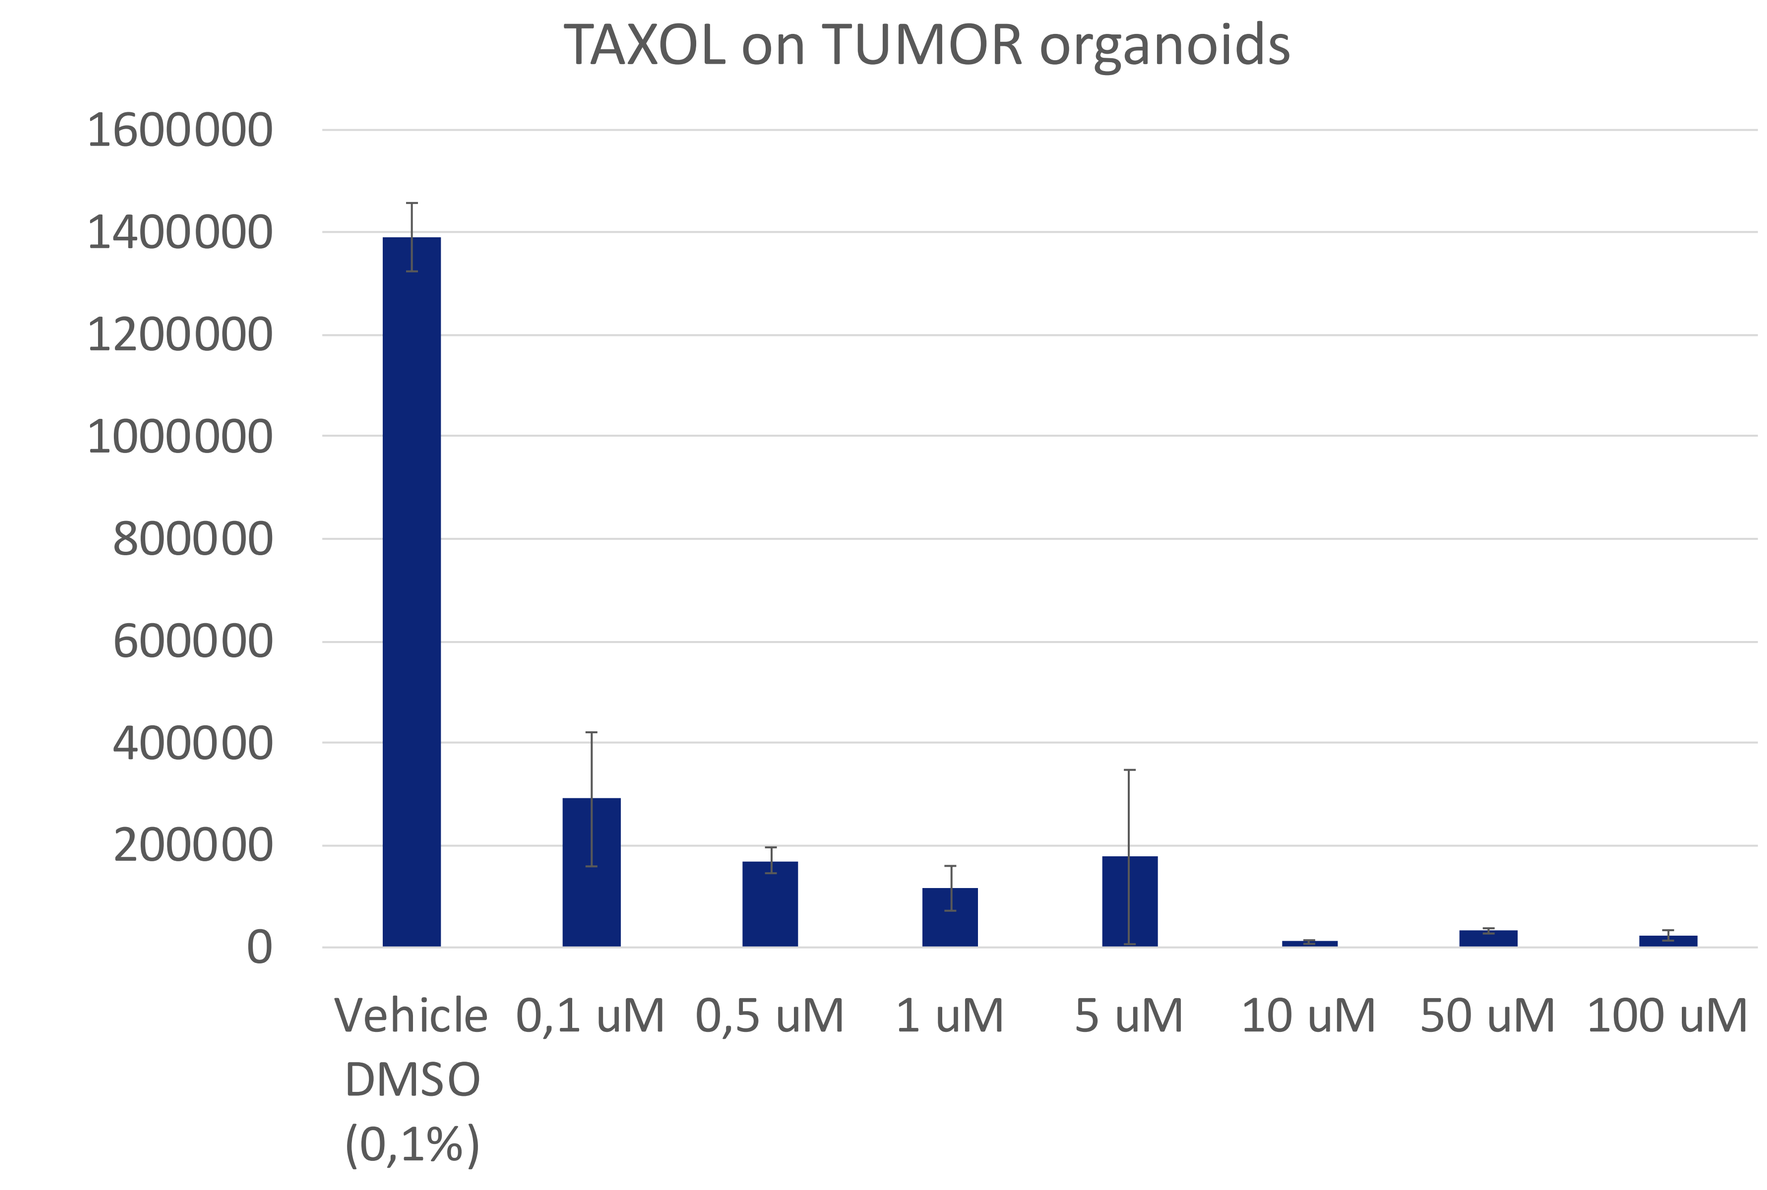
TAXOL on TUMOR organoids
1600000
1400000
1200000
1000000
800000
600000
400000
200000
0
Vehicle DMSO (0,1%)
0,1 uM
0,5 uM
1 uM
5 uM
10 uM
50 uM
100 uM

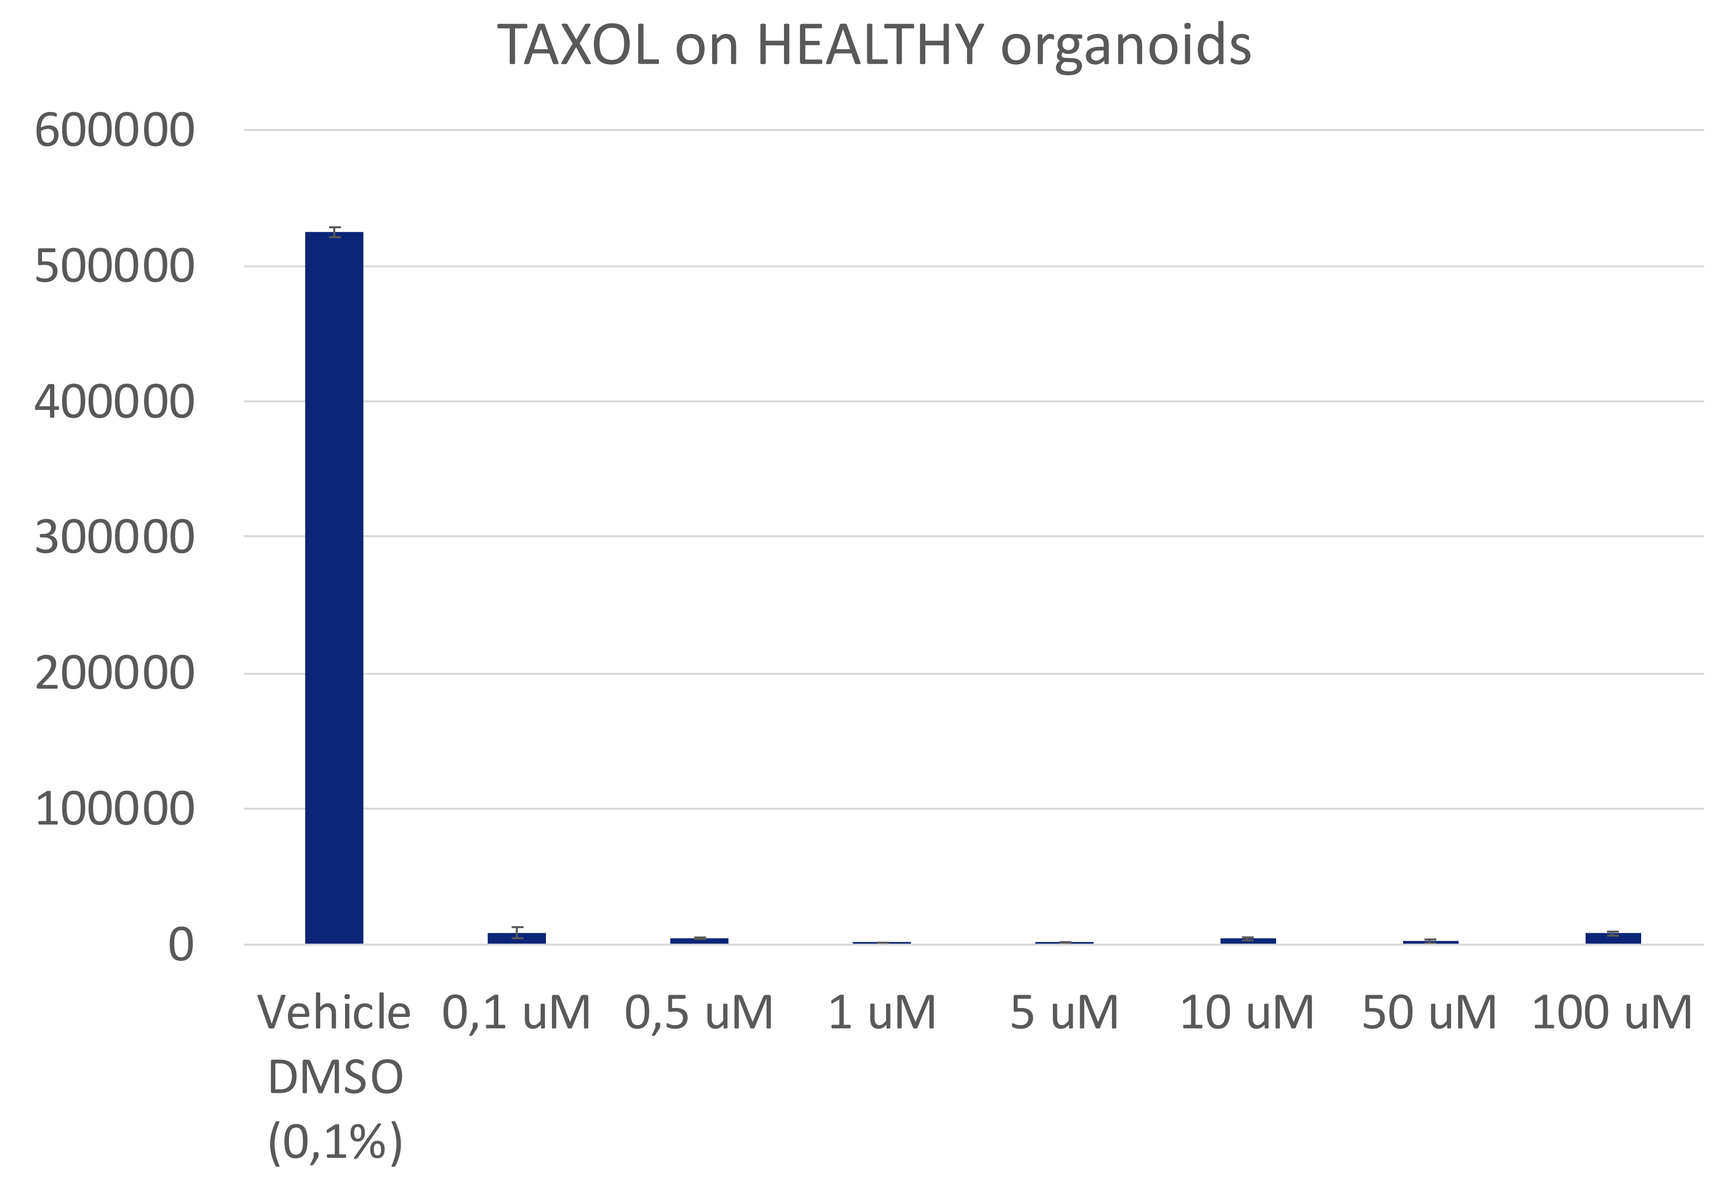
TAXOL on HEALTHY organoids
600000
500000
400000
300000
200000
100000
0
Vehicle DMSO (0,1%)
0,1 uM
0,5 uM
1 uM
5 uM
10 uM
50 uM
100 uM

- PDOs recapitulate the histological and morphological features of the primary tumor
- CD45+ cells remain in PDOs (day 4)
- Drug assessment of taxol (chemotherapy regimen) on PDOs showed a dose-response curve on tumoral PDOs and a lot of toxicity on healthy PDOs as well.

Discussion

A limit often described in cancer 3D models is the lack of cancer tumor microenvironment, and particularly of non-immune cells, such as stromal cells and immune cells[2]. With the new era of immune checkpoint inhibitors, a co-culture system with immune-infiltrating cells would be better to study their efficacy. PDOs associated in a microfluidic device may be more accurate to assess drugs in different modes of administration because of their physiological relevance with the primary tumor.

REFERENCES

1 Burotto et al. (2014) Biomarkers in early-stage non-small-cell lung cancer: current concepts and future directions. J Thorac Oncol. 9:1609-17.
2 Kim et al. (2019) Patient-derived lung cancer organoids as in vitro cancer models for therapeutic screening. Nature Commun. 10:1–15

CONCLUSION

The challenge is to develop more relevant *in vitro* models, recapitulating histological and genetic features of lung cancer taking into consideration the tumor microenvironment and its various cells component. The vascular and immune components must now be included in the PDO system, as they largely influence tumor growth and response to drugs. This will be of high importance in the perspective of personalized treatment and combination treatment. Lung cancer has been shown to be highly immunogenic, and is thus responsive to checkpoint blockade therapy. Oncolytic virotherapy is an emergent therapy for the treatment of cancer that could advantageously combine with existing treatments. New oncolytic virus candidates will be evaluated on this innovative device.

EUROoCs– Uppsala, Sweden– July, 2020