BT-001, an oncolytic vaccina virus armed with a Treg-depleting anti-CTLA-4 antibody and GM-CSF to target the tumor microenvironment

Monika Semmrich [1], Jean-Baptiste Marchand [2], Laetitia Fend [2], Matilda Rehn [1], Nathalie Silvestre [2],
Johann Foloppe [2], Linda Mårtensson [1], Ingrid Teige [1], Eric Quéméneur [2], Björn Frendéus[1]

[1] BioInvent International AB, Lund, Sweden, [2] Transgene S.A., Illkirch-Graffenstaden, France

Abstract #594

1. Background

Treatment with checkpoint inhibitor antibodies results in long-lasting antitumor responses in a variety of cancers [1]. However, only a small fraction of patients responds to the treatment, probably due to inadequate tumor infiltration with immune cells. While combination therapy with anti-CTLA-4 and anti-PD-1 antibodies significantly improves efficacy, concerns with tolerability has limited wide-spread clinical use [2].

Here we present a potentially safe and more efficacious strategy to combine anti-CTLA-4 and anti- PD-1/PDL1 checkpoint inhibition in the context of oncolytic virotherapy. A Treg-depleting anti-CTLA-4 antibody has been vectorized alongside GM-CSF into the Invir.IO™ oncolytic Vaccinia virus (oVV) based platform. This product named BT-001 consists of the Copenhagen oVV strain - deleted in J2R and I4L viral genes allowing restricted replication in proliferating cells - and the human CTLA-4-specific antibody 4-E03 IgG1, which shows improved Treg-depletion compared with ipilimumab.

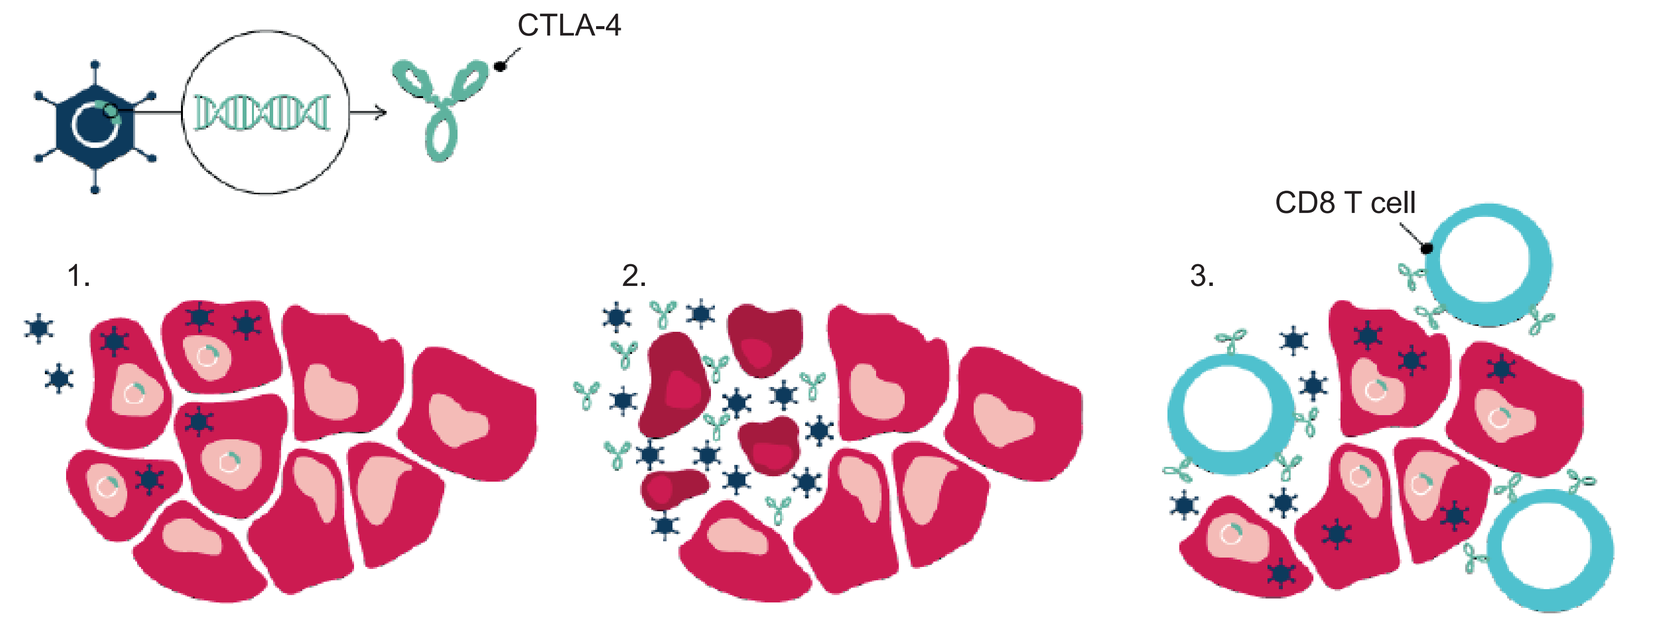
CTLA-4
CD8 T cell
1.
2.
3.

Figure 1. Local administration of BT-001 combines oncolytic activities with high intratumoral concentrations of anti-CTLA-4 antibody and GM-CSF eliciting a stronger antitumor response with improved safety profile.

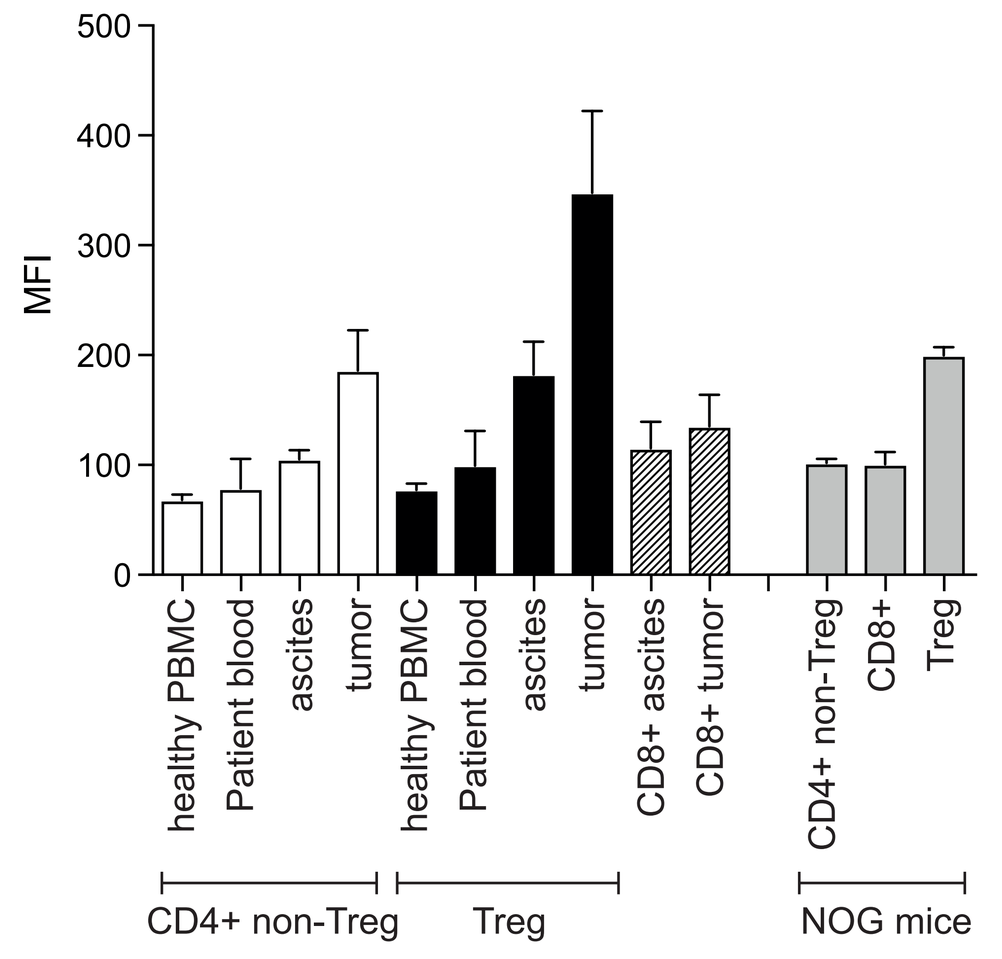
MFI
healthy PBMC
Patient blood
ascites
tumor
CD8+ ascites
CD8+ tumor
CD4+ non-Treg
CD8+
Treg
CD4+ non-Treg
Treg
NOG mice

Figure 2. In primary patient material, CTLA-4 is highest expressed on intratumoral Treg cells which makes them a good target for Treg depleting antibodies.

2. Generation of BT-001 and mouse surrogate mBT-001

Figure 3. Treg depleting and blocking anti-CTLA-4 mAb 4-E03

BioInvent's F.I.R.S.T. discovery platform [3] was used to isolate scFv antibody fragments recognizing human or mouse CTLA-4. Target-specific antibody clones were classified as actives, transferred to full-length IgG format, and further characterized biochemically and functionally. The antibody clone 4-E03 was chosen as candidate for vectorization in Copenhagen oVV. It blocks CD80/CTLA-4 and CD86/CTLA-4 interactions with the same potency as ipilimumab (a) but the Treg depleting activity is improved compared to ipilimumab as demonstrated in a PBMC-NOG/SCID transfer model in vivo (b). An anti-mouse CTLA-4 antibody (clone 5-B07) with similar functional effects in ligand blocking and Treg depletion (data not shown) was selected for the generation of the murine surrogate virus mBT-001.

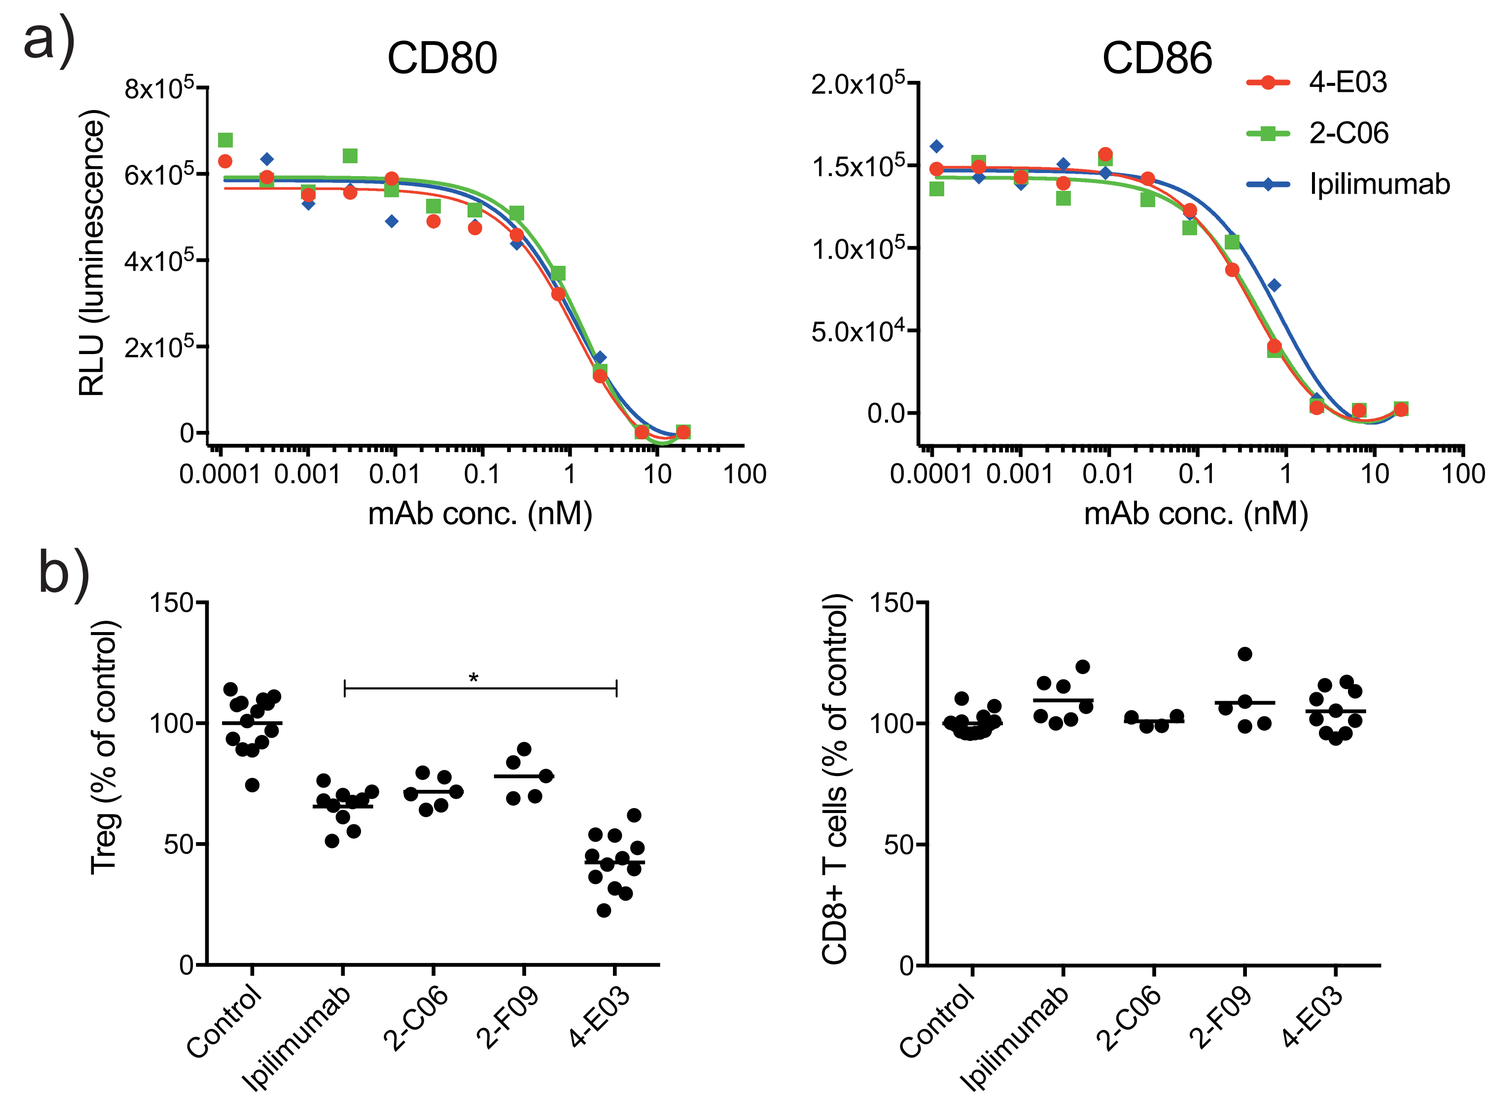
a)
CD80
CD86
RLU (luminescence)
mAb conc. (nM)
4-E03
2-C06
Ipilimumab
b)
Treg (% of control)
CD8+ T cells (% of control)
Control
Ipilimumab
2-C06
2-F09
4-E03

Figure 4. Oncolytic Vaccinia virus Copenhagen strain

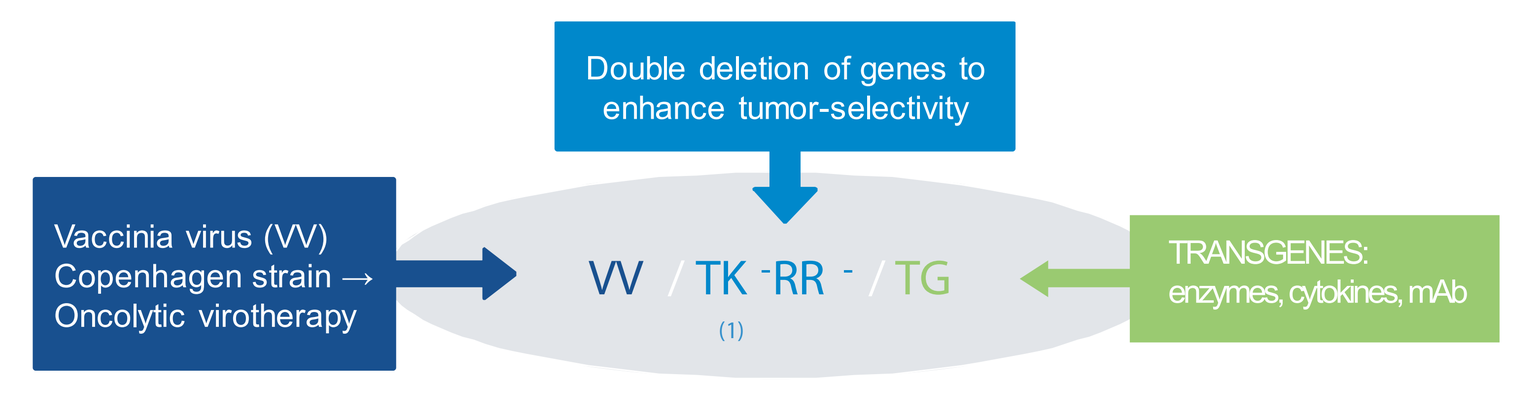
Double deletion of genes to enhance tumor-selectivity
Vaccinia virus (VV) Copenhagen strain → Oncolytic virotherapy
VV / TK⁻RR⁻ / TG
TRANSGENES: enzymes, cytokines, mAb

Copenhagen oVV strain
- is deleted in J2R (TK locus) and I4L (RR locus) viral genes involved in nucleotide synthesis to restrict replication in proliferating cells
- allows large DNA insertions with successful vectorization of various expression cassettes
- has the best oncolytic activity among VACV strains
- induces Immune Cell Death

Figure 5. Vaccinia virus expressing 4-E03 and GM-CSF (BT-001) or 5-B07 and mGM-CSF (mBT-001)

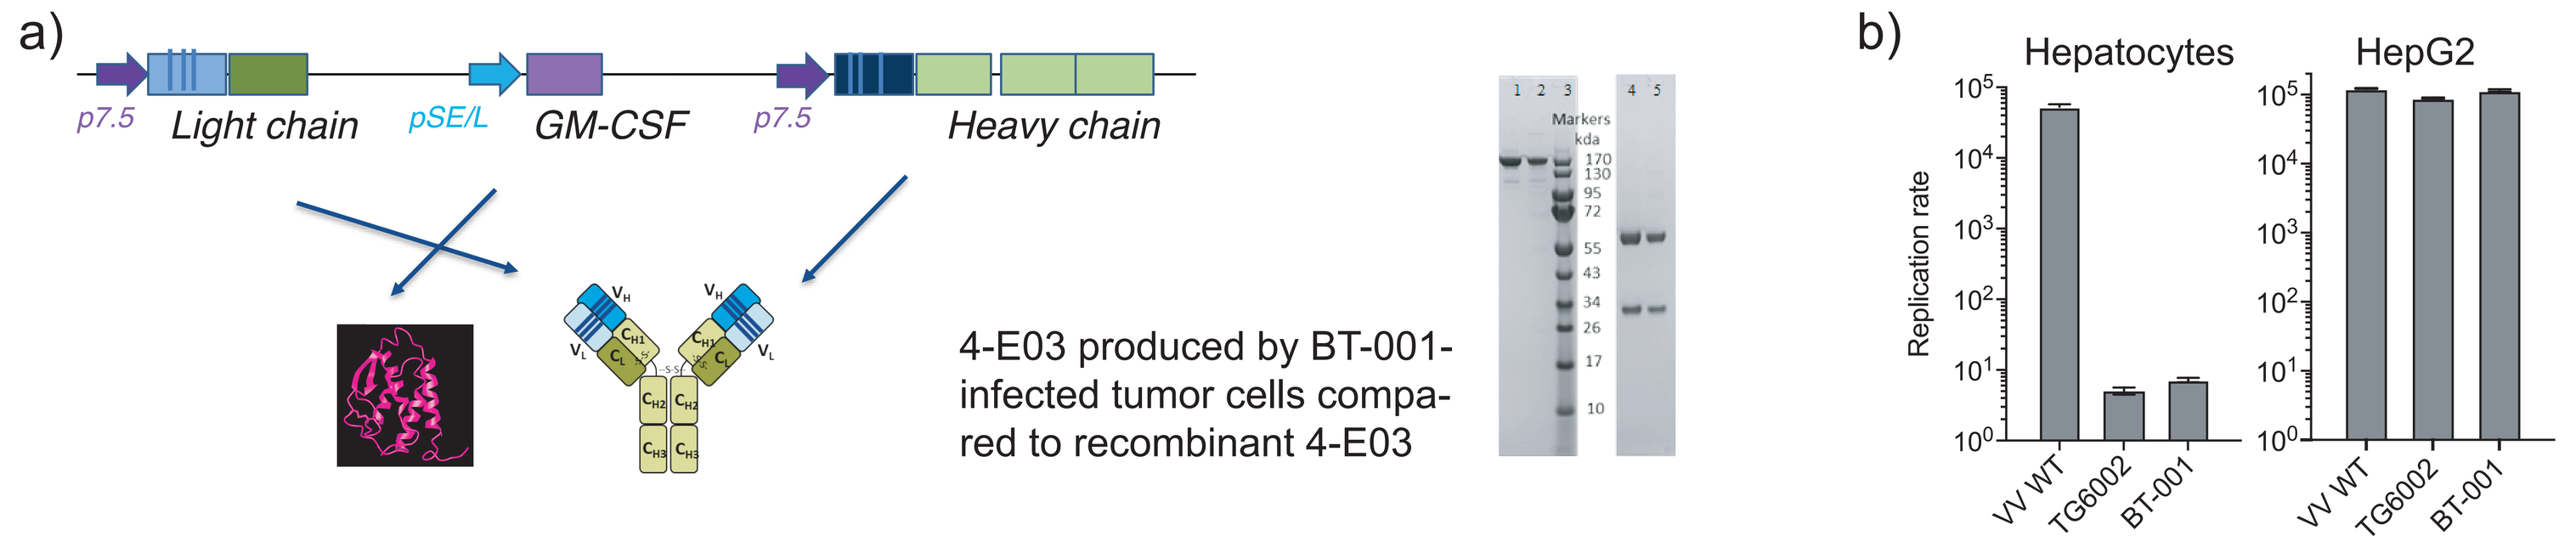
a)
p7.5
Light chain
pSE/L
GM-CSF
p7.5
Heavy chain
4-E03 produced by BT-001-infected tumor cells compared to recombinant 4-E03
b)
Hepatocytes
HepG2
Replication rate
VV WT
TG6002
BT-001

a) Anti-CTLA-4 mAb and GM-CSF were vectorized in Copenhagen Vaccinia virus. Thereby deleted J2R and I4L genes were replaced by heavy and light chain of anti-CTLA-4, respectively. The expression cassette encoding GM-CSF was also placed at the I4L locus.
b) BT-001 selectively replicated in tumor cells and not in normal cells, similar to the clinically validated oVV TG6002.

3. Transgenes functionality and kinetics

Figure 6. Transgenes produced by virus infected tumor cells are funtional

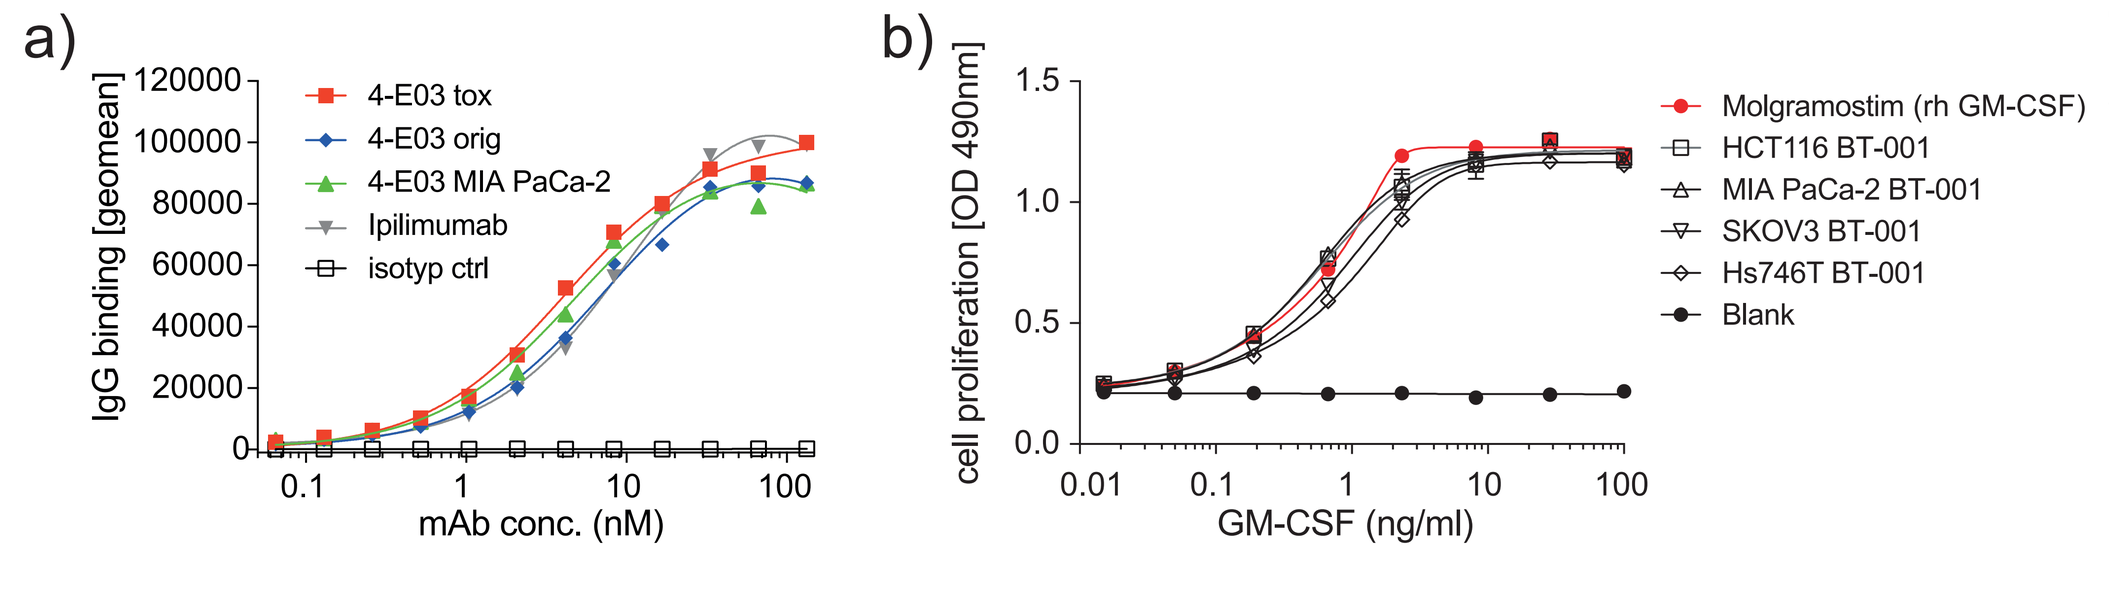
a)
IgG binding [geomean]
mAb conc. (nM)
4-E03 tox
4-E03 orig
4-E03 MIA PaCa-2
Ipilimumab
isotyp ctrl
b)
cell proliferation [OD 490nm]
GM-CSF (ng/ml)
Molgramostim (rh GM-CSF)
HCT116 BT-001
MIA PaCa-2 BT-001
SKOV3 BT-001
Hs746T BT-001
Blank

Transgenes GM-CSF and αCTLA-4 produced from virally infected MIA PaCa-2 cells were compared to recombinantly produced antibody and GM-CSF.
a) Recombinant and virally produced 4-E03 showed identical binding to CTLA-4 expressing cells. b) Virally produced GM-CSF induced GM-CSF dependent cell proliferation to the same extent as recombinant GM-CSF.

Figure 7. Vectorization allows intra-tumoral accumulation of virus and transgenes with low systemic exposure

Amount of transgenes in tumor vs blood was measured for both anti-CTLA-4 mAb and GM-CSF (not shown) in a) xenograft and b) syngeneic CT26 model.

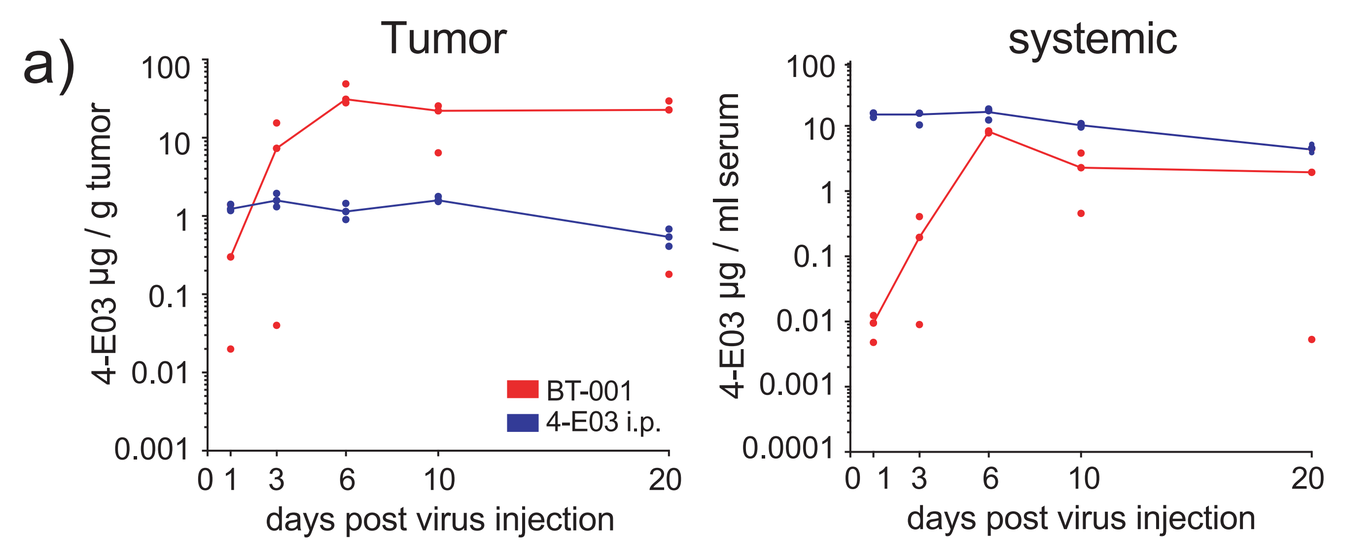
a)
Tumor
systemic
4-E03 µg / g tumor
4-E03 µg / ml serum
BT-001
4-E03 i.p.
days post virus injection

a) Concentration of anti-human CTLA-4 mAb 4-E03 was monitored in Xenograft tumor model. LoVo human colorectal tumor cells were implanted subcutaneously to Nude mice and 10^5 PFU of BT-001 was injected i.t. One injection of 4-E03 i.p. at 3 mg/kg was used as benchmark.

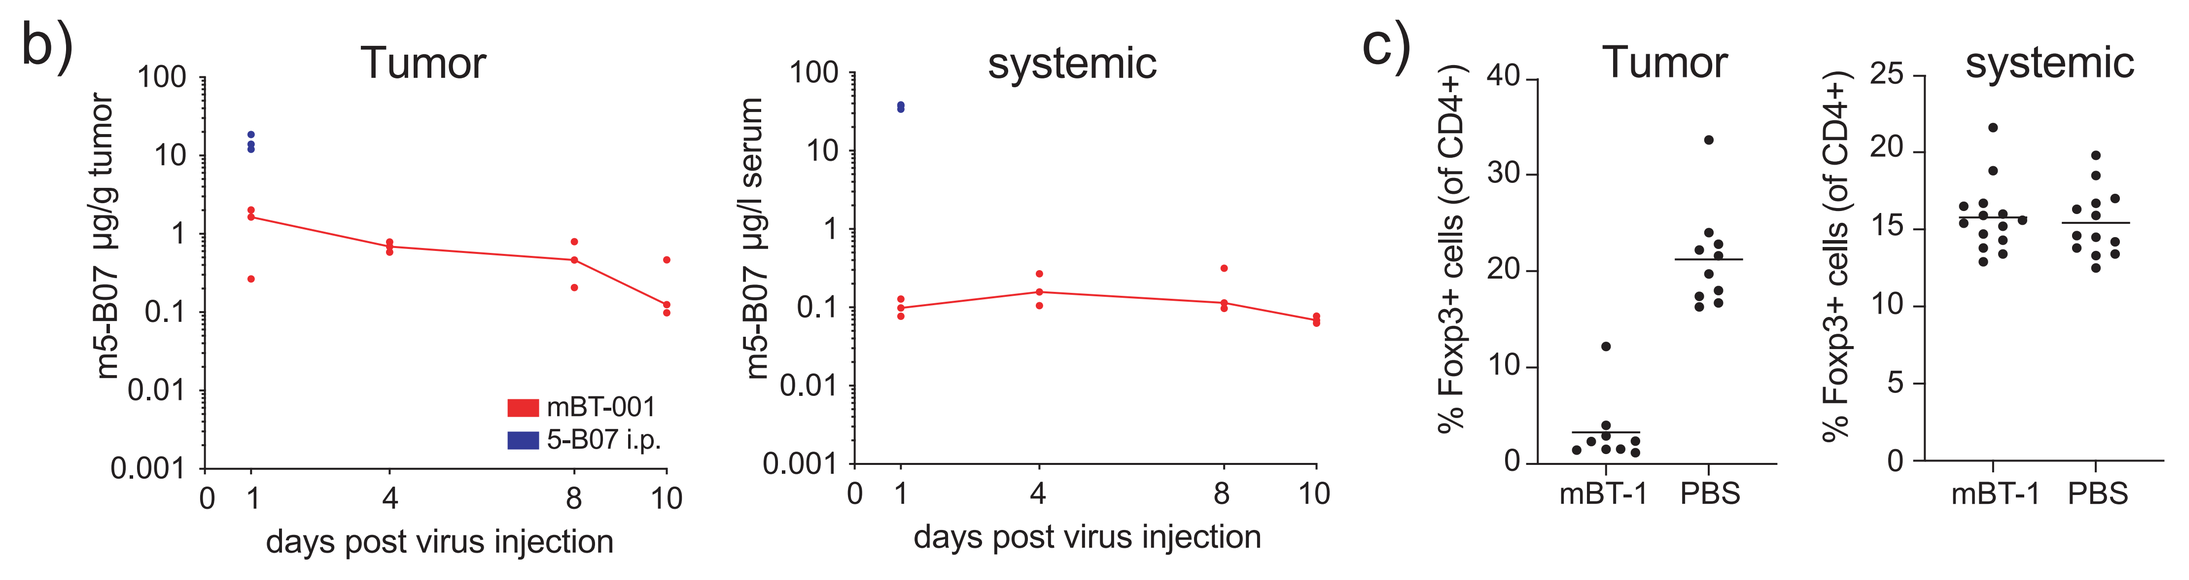
b)
Tumor
systemic
m5-B07 µg/g tumor
m5-B07 µg/l serum
mBT-001
5-B07 i.p.
days post virus injection
c)
Tumor
systemic
% Foxp3+ cells (of CD4+)
mBT-1
PBS

b) Concentration of anti-mouse CTLA-4 mAb 5-B07 was monitored in syngeneic CT26 tumor model. CT26 colorectal tumor cells were implanted subcutaneously to Balb/c mice and virus was injected 3 times i.t. One injection intraperitoneally of 5-B07 at 3 mg/kg was used as benchmark.

c) Intratumoral administration of mBT-001 was associated with sustained intratumoral antibody exposure and intratumoral Treg depletion, systemic exposure was orders of magnitude lower and not associated peripheral Treg depletion.

4. Anti-tumor activity in vivo of mBT-001

Figure 8. Improved survival after treatment with mBT-001 in several syngeneic tumor models

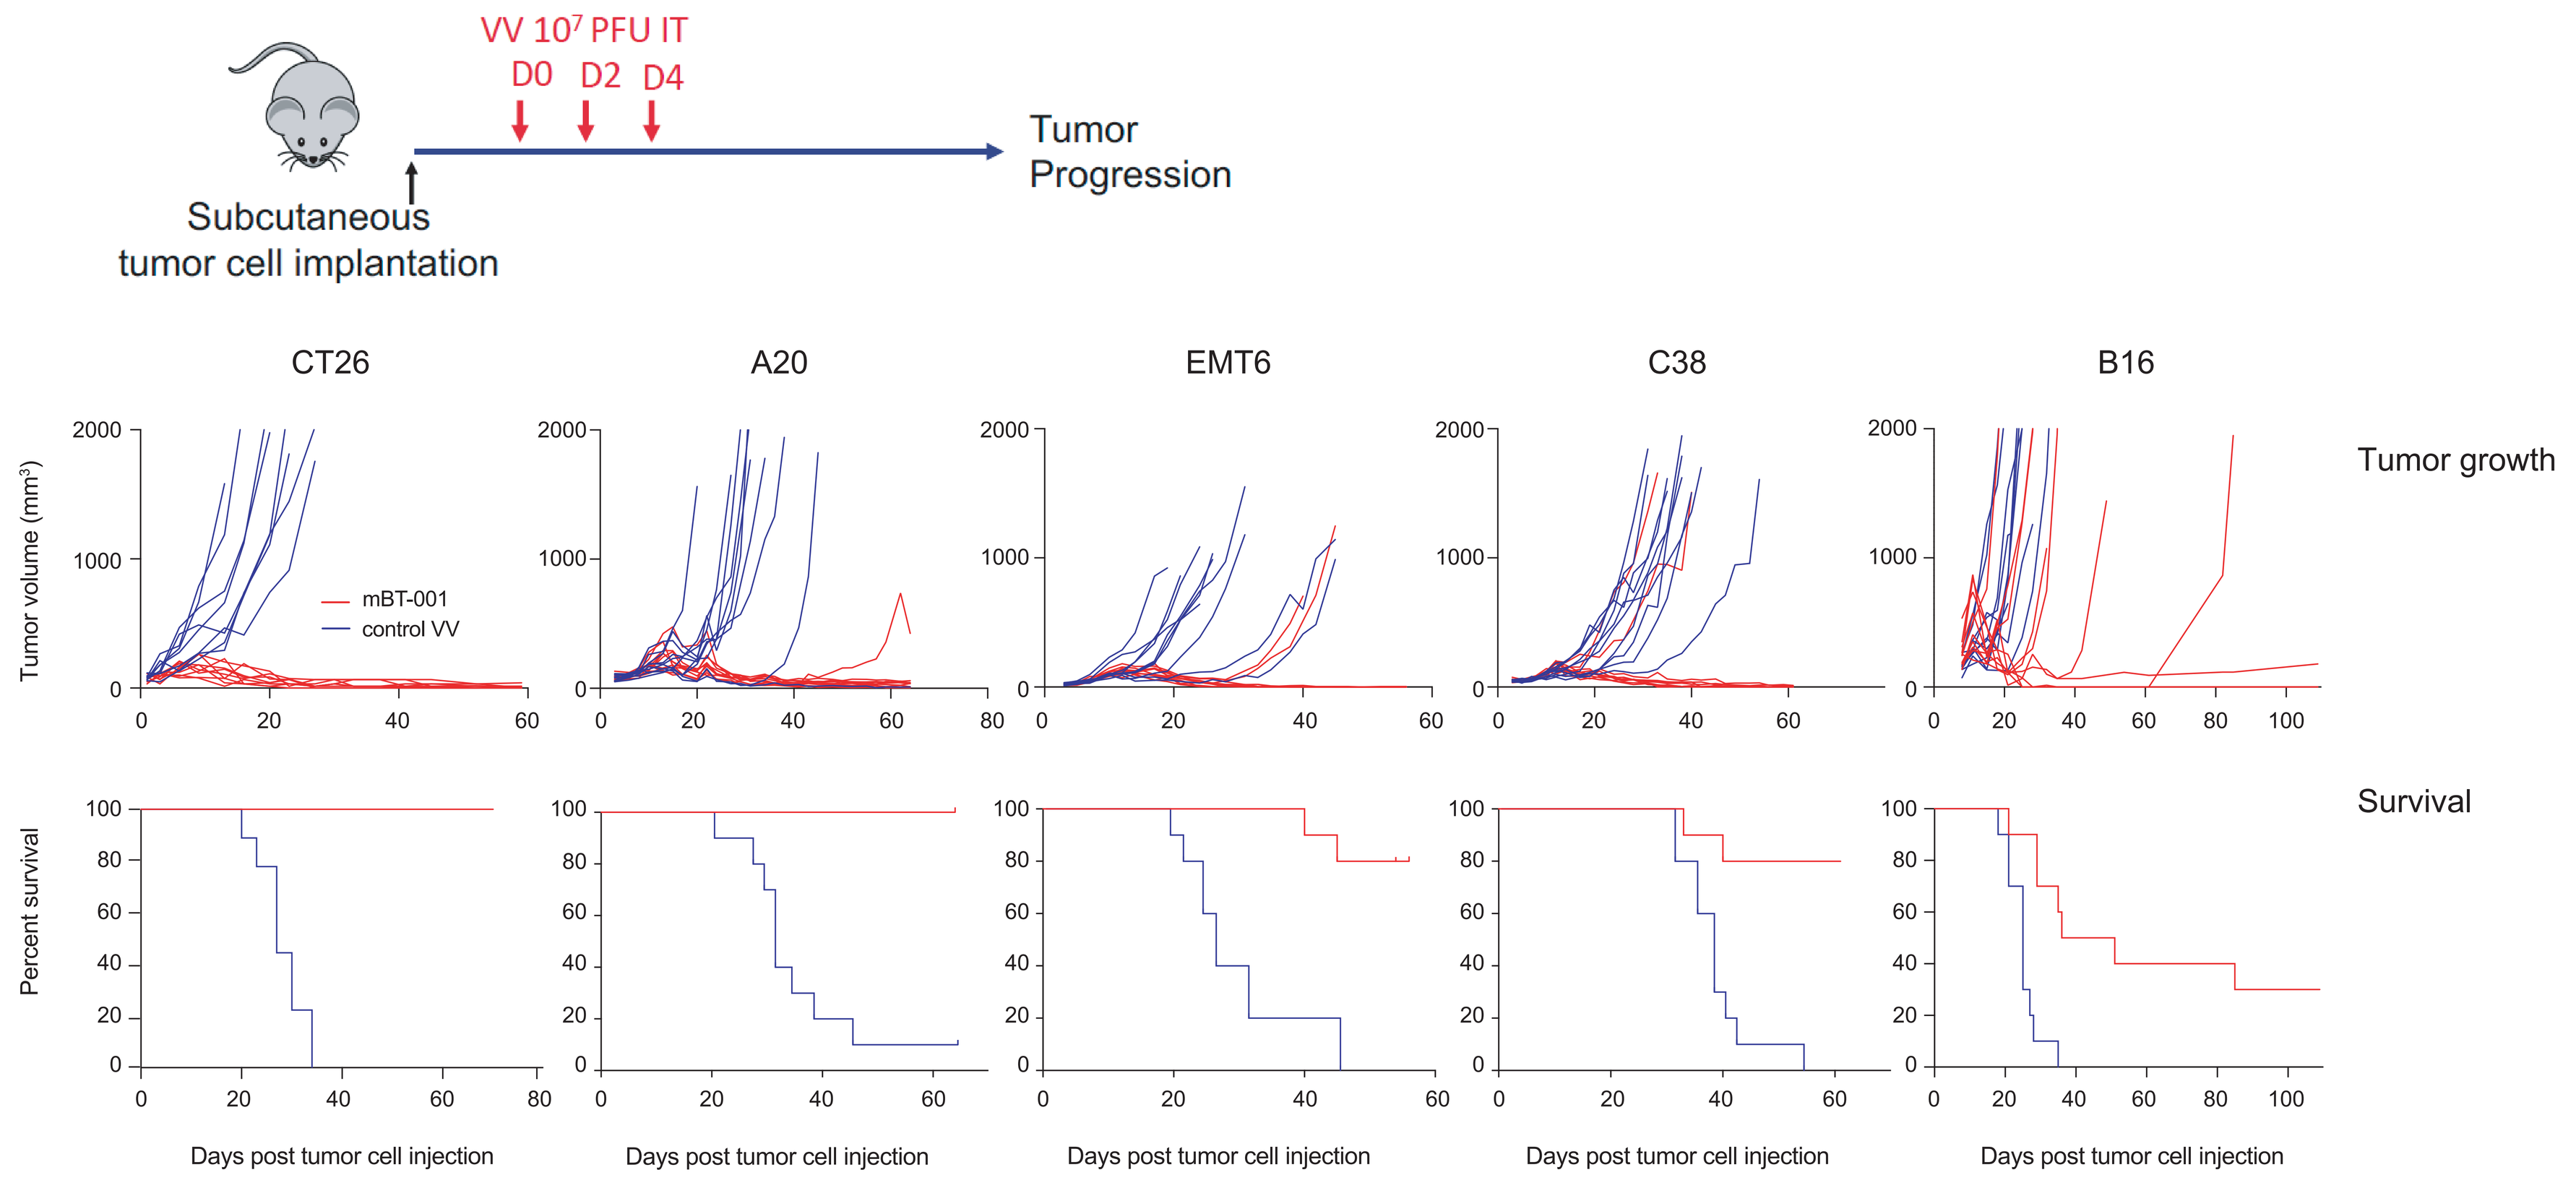
VV 10⁷ PFU IT
D0 D2 D4
Subcutaneous tumor cell implantation
Tumor Progression
CT26
A20
EMT6
C38
B16
Tumor growth
Tumor volume (mm³)
mBT-001
control VV
Survival
Percent survival
Days post tumor cell injection

Antitumoral activity of oncolytic Copenhagen vaccinia virus encoding an anti-mouse CTLA-4 and murine GM-CSF (mBT-001) was assessed in different syngeneic tumor models and after 3 i.t. administrations of 10^7 PFU. Results demonstrate broad and potent antitumor activity of mBT-001 in immune inflamed and immune excluded models.

Figure 9. Intratumoral injection of mBT-001 induces systemic antitumor immunity (abscopal effect)

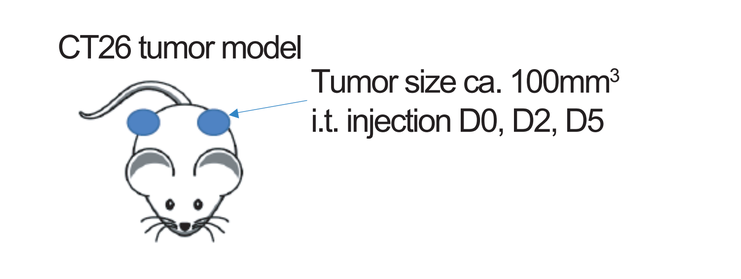
CT26 tumor model
Tumor size ca. 100mm³
i.t. injection D0, D2, D5

Local, intratumoral administration of mBT-001 induced complete tumor regression in contralateral uninjected tumors in 8/10 mice demonstrating the induction of systemic antitumor responses, or the abscopal effect. There was no evidence of viral particle dissemination to uninjected tumors as assessed by plaque assay (data not shown).

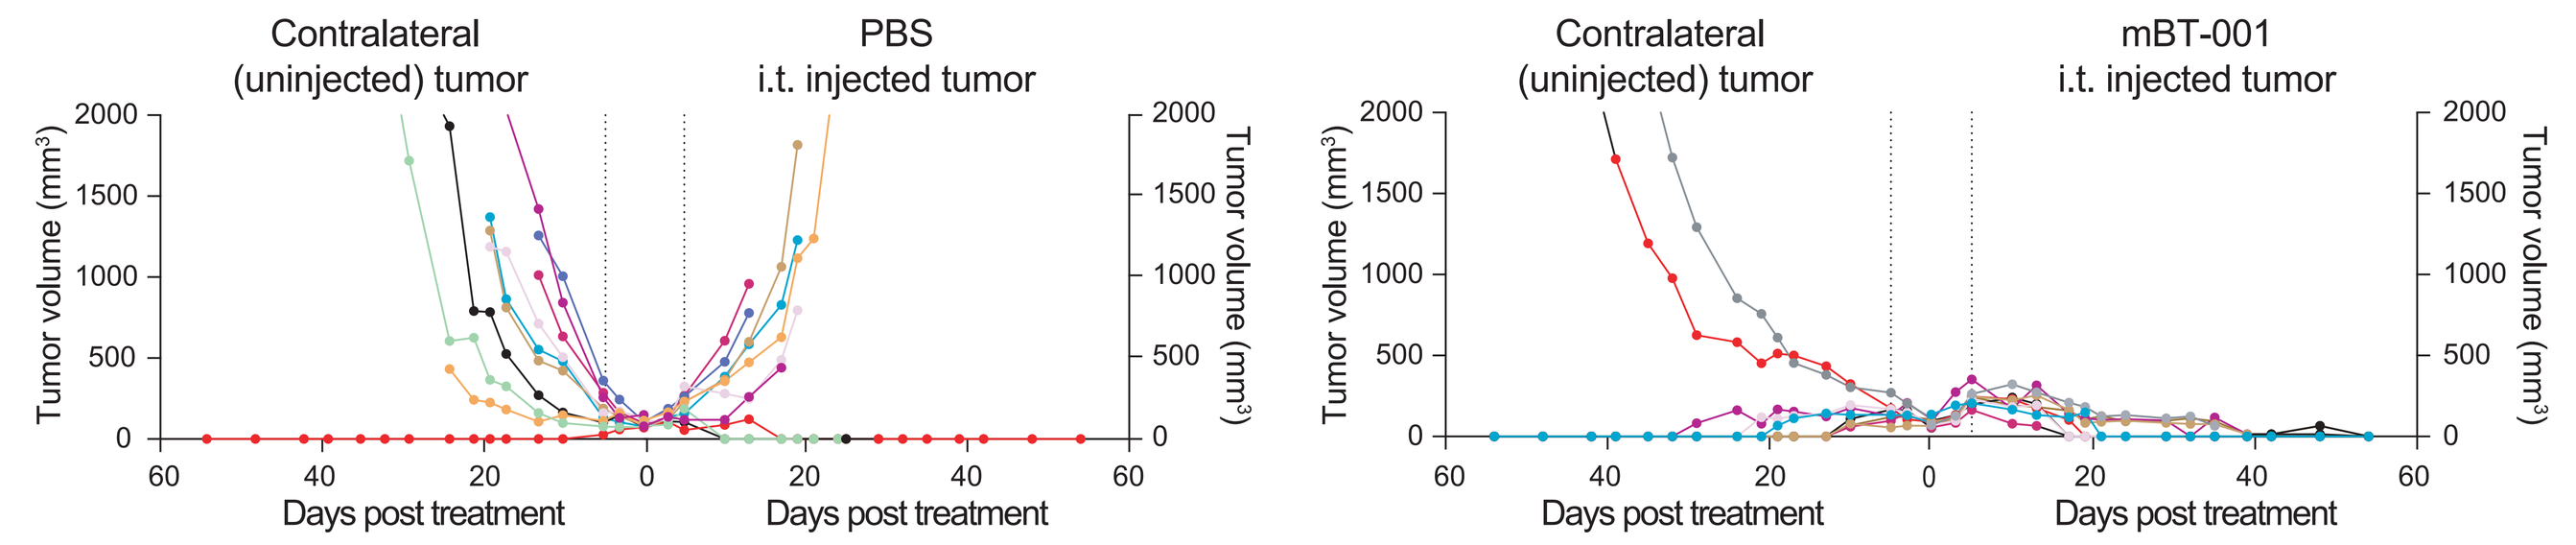
Contralateral (uninjected) tumor
PBS i.t. injected tumor
Contralateral (uninjected) tumor
mBT-001 i.t. injected tumor
Tumor volume (mm³)
Days post treatment

Figure 10. mBT-001 does induce memory responses

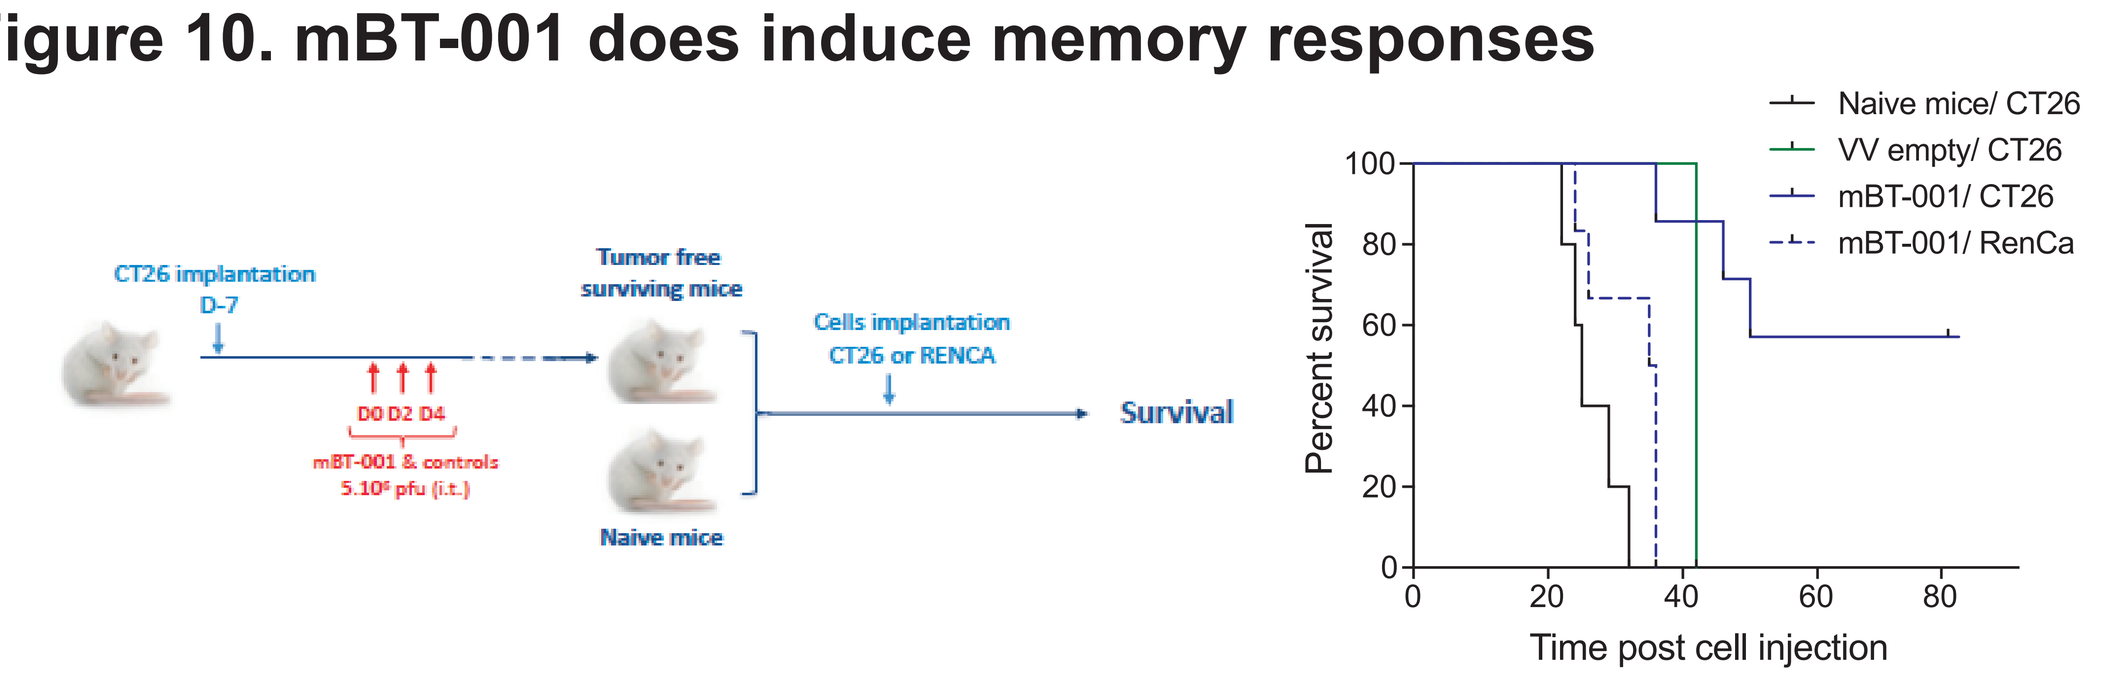
CT26 implantation D-7
D0 D2 D4
mBT-001 & controls
Tumor free surviving mice
Naive mice
Cells implantation CT26 or RENCA
Survival
Naive mice/ CT26
VV empty/ CT26
mBT-001/ CT26
mBT-001/ RenCa
Percent survival
Time post cell injection

Animals cured by intratumoral treatment with mBT-001 were signinficantly protected from rechallange with the same but not unrelated tumor cells.

Figure 11. Anti-tumor response is CD8 T cell dependent

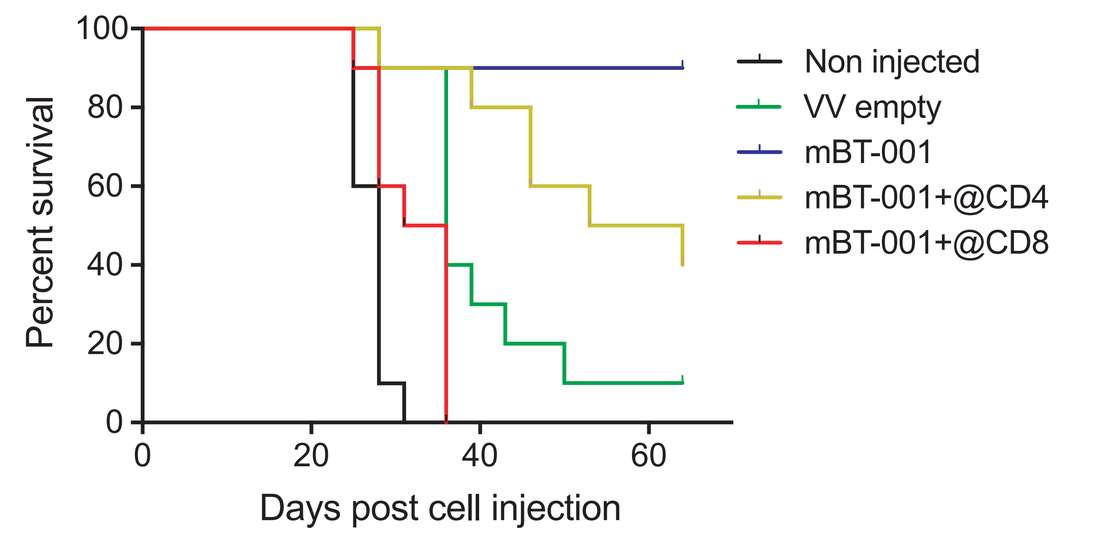
Non injected
VV empty
mBT-001
mBT-001+@CD4
mBT-001+@CD8
Percent survival
Days post cell injection

Depletion of CD8+ and CD4+ T cells prior to treament of CT26-bearing mice with mBT-001 demonstrated a crucial role for CD8+ T cells in the anti-tumor response. The dependency on CD4+ T cells was less significant.

Figure 12. Combination of mBT-001 with anti-PD-1 is benificial

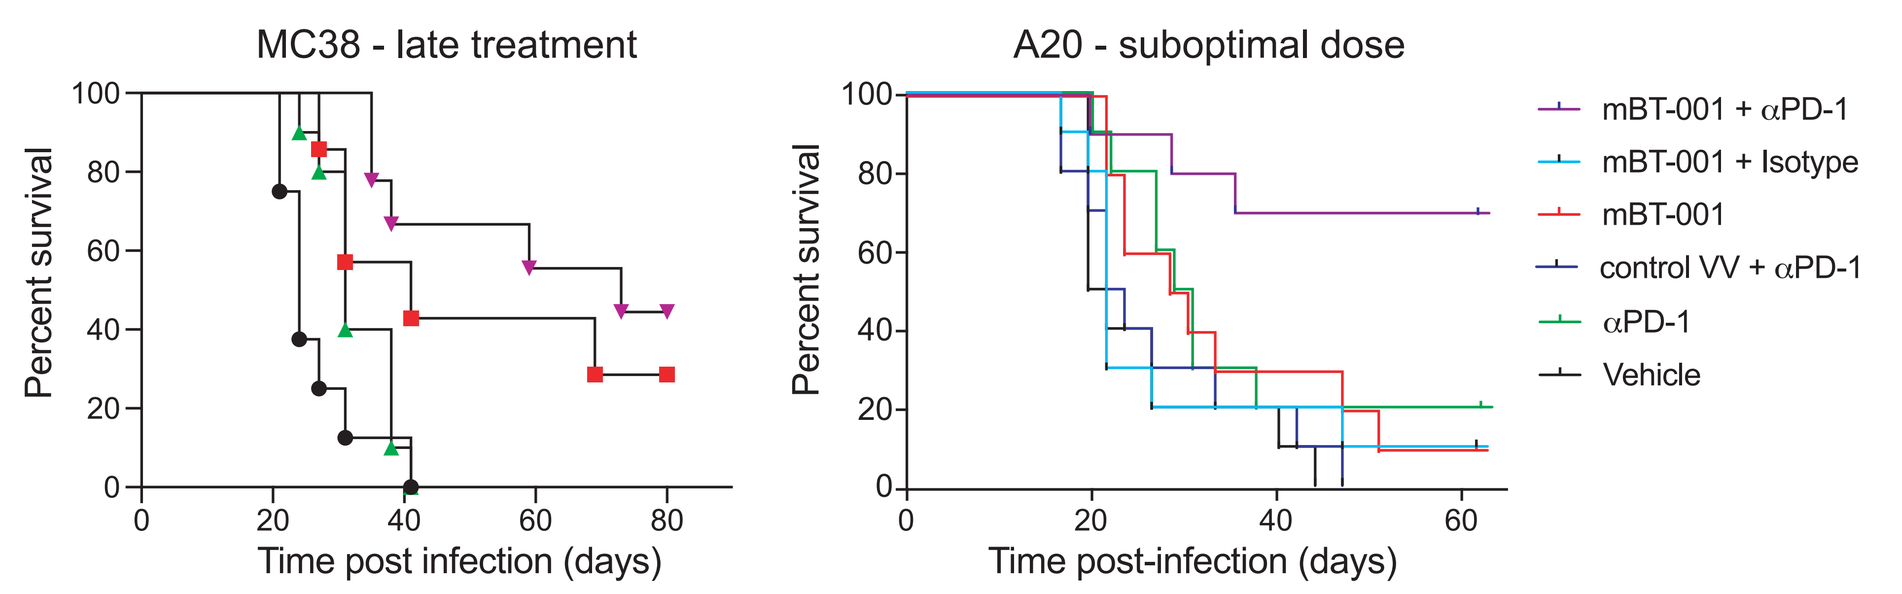
MC38 - late treatment
A20 - suboptimal dose
Percent survival
Time post infection (days)
Time post-infection (days)
mBT-001 + αPD-1
mBT-001 + Isotype
mBT-001
control VV + αPD-1
αPD-1
Vehicle

The potential benefit of the combination of mBT-001 and anti-PD-1 was evaluated either by testing a sub-optimal dose of mBT-1 (10^5 PFU) or by starting the treatment late, when the tumors have already reached 200mm³.

5. Conclusions

- BT-001 is a multifunctional oVV co-developed by Transgene and BioInvent that encodes a Treg - depleting anti-CTLA-4 antibody as well as the cytokine GM-CSF.
- BT-001 combines potent oncolytic activities with production of high intratumoral concentrations of anti-CTLA-4 antibody and GM-CSF, and very low systemic exposure.
- The murine surrogate mBT-001 has demonstrated an outstanding antitumoral activity in several syngeneic tumor models. This antitumoral activity is further enhanced by a combination with anti-PD-1 treatment and leads to systemic and long-lasting antitumoral immune responses.
- It is expected to enter a phase I clinical trial before the end of 2020.

6. References

1. Sharma, P. and J.P. Allison, Science, 2015. 348(6230): p. 56-61.
2. Postow, M.A., et al., N Engl J Med, 2015. 372(21): p. 2006-17.
3. Soderlind, E., et al., Nat Biotechnol, 2000. 18(8): p. 852-6.
4. Arce Vargas, F., et al., Cancer Cell, 2018.
5. Kleinpeter, P., et al., Oncoimmunology, 2016. 5(10): p. e1220467.
6. Fend, L., et al., Cancer Res, 2017. 77(15): p. 4146-4157.
7. www.transgene.fr, www.bioinvent.se