Extracellular vesicles (EV) - mediators of therapeutic vaccination?

In vivo and in vitro characterization of EVs generated after infection of human and murine cells with therapeutic poxviruses

Lucas Walther[1,2], Caroline Tosch[1], Jules Deforges[1], Christine Carapito[3], Magali Rompais[3], Marie-Christine Claudepierre[1], Christelle Remy[1], Nathalie Silvestre[1], Kuang-Jing HUANG[2,4], Kaidre Bendjama[1], Eric Quemeneur[1], Jacky G. Goetz[2], Vincent Hyenne[2], Karola Rittner[1]

[1] **Transgene** SA, Illkirch-Graffenstaden, France. [2] **Tumor Biomechanics Lab, INSERM UMR_S1109,** Université de Strasbourg, Fédération de Médecine Translationnelle de Strasbourg (FMTS), Equipe Labellisée Ligue contre le Cancer, Strasbourg, France. [3] **Laboratoire de Spectrométrie de Masse Bio-Organique, Institut Pluridisciplinaire Hubert Curien, UMR 7178**, CNRS-Université de Strasbourg, Strasbourg, France. [4] **Department of Integrated Structural Biology, Institut de Génétique et de Biologie Moléculaire et Cellulaire (IGBMC)**, 67404, Illkirch, France.

Contact: rittner@transgene.fr

AACR 2023
Abstract # 697
Poster # 17

INTRODUCTION

Poxviral vectors are among the most promising therapeutic vaccines, demonstrated in ongoing clinical trials: TG4001, a MVA encoding the HPV oncoviral antigens E6 and E7 applied in HPV+ anogenital cancers (AACR poster CT045) and TG4050, a personalized MVA vaccine against ovarian and head and neck cancer (AACR poster LB205).

All cells, but especially virus-infected cells, communicate with their environment by secreting extracellular vesicles (EVs). In this work, we inquired whether EVs played a role in the therapeutic vaccination process with MVA vaccines:

- **How can we separate EVs from poxviruses after infection** (PBMCs or murine DC2.4 cells) **?**
- **How does poxvirus infection modulate the secretion and content of EVs ?**
- **Are EVs immunogenic ?**
- **Do they mediate therapeutic effects ?**

PROJECT

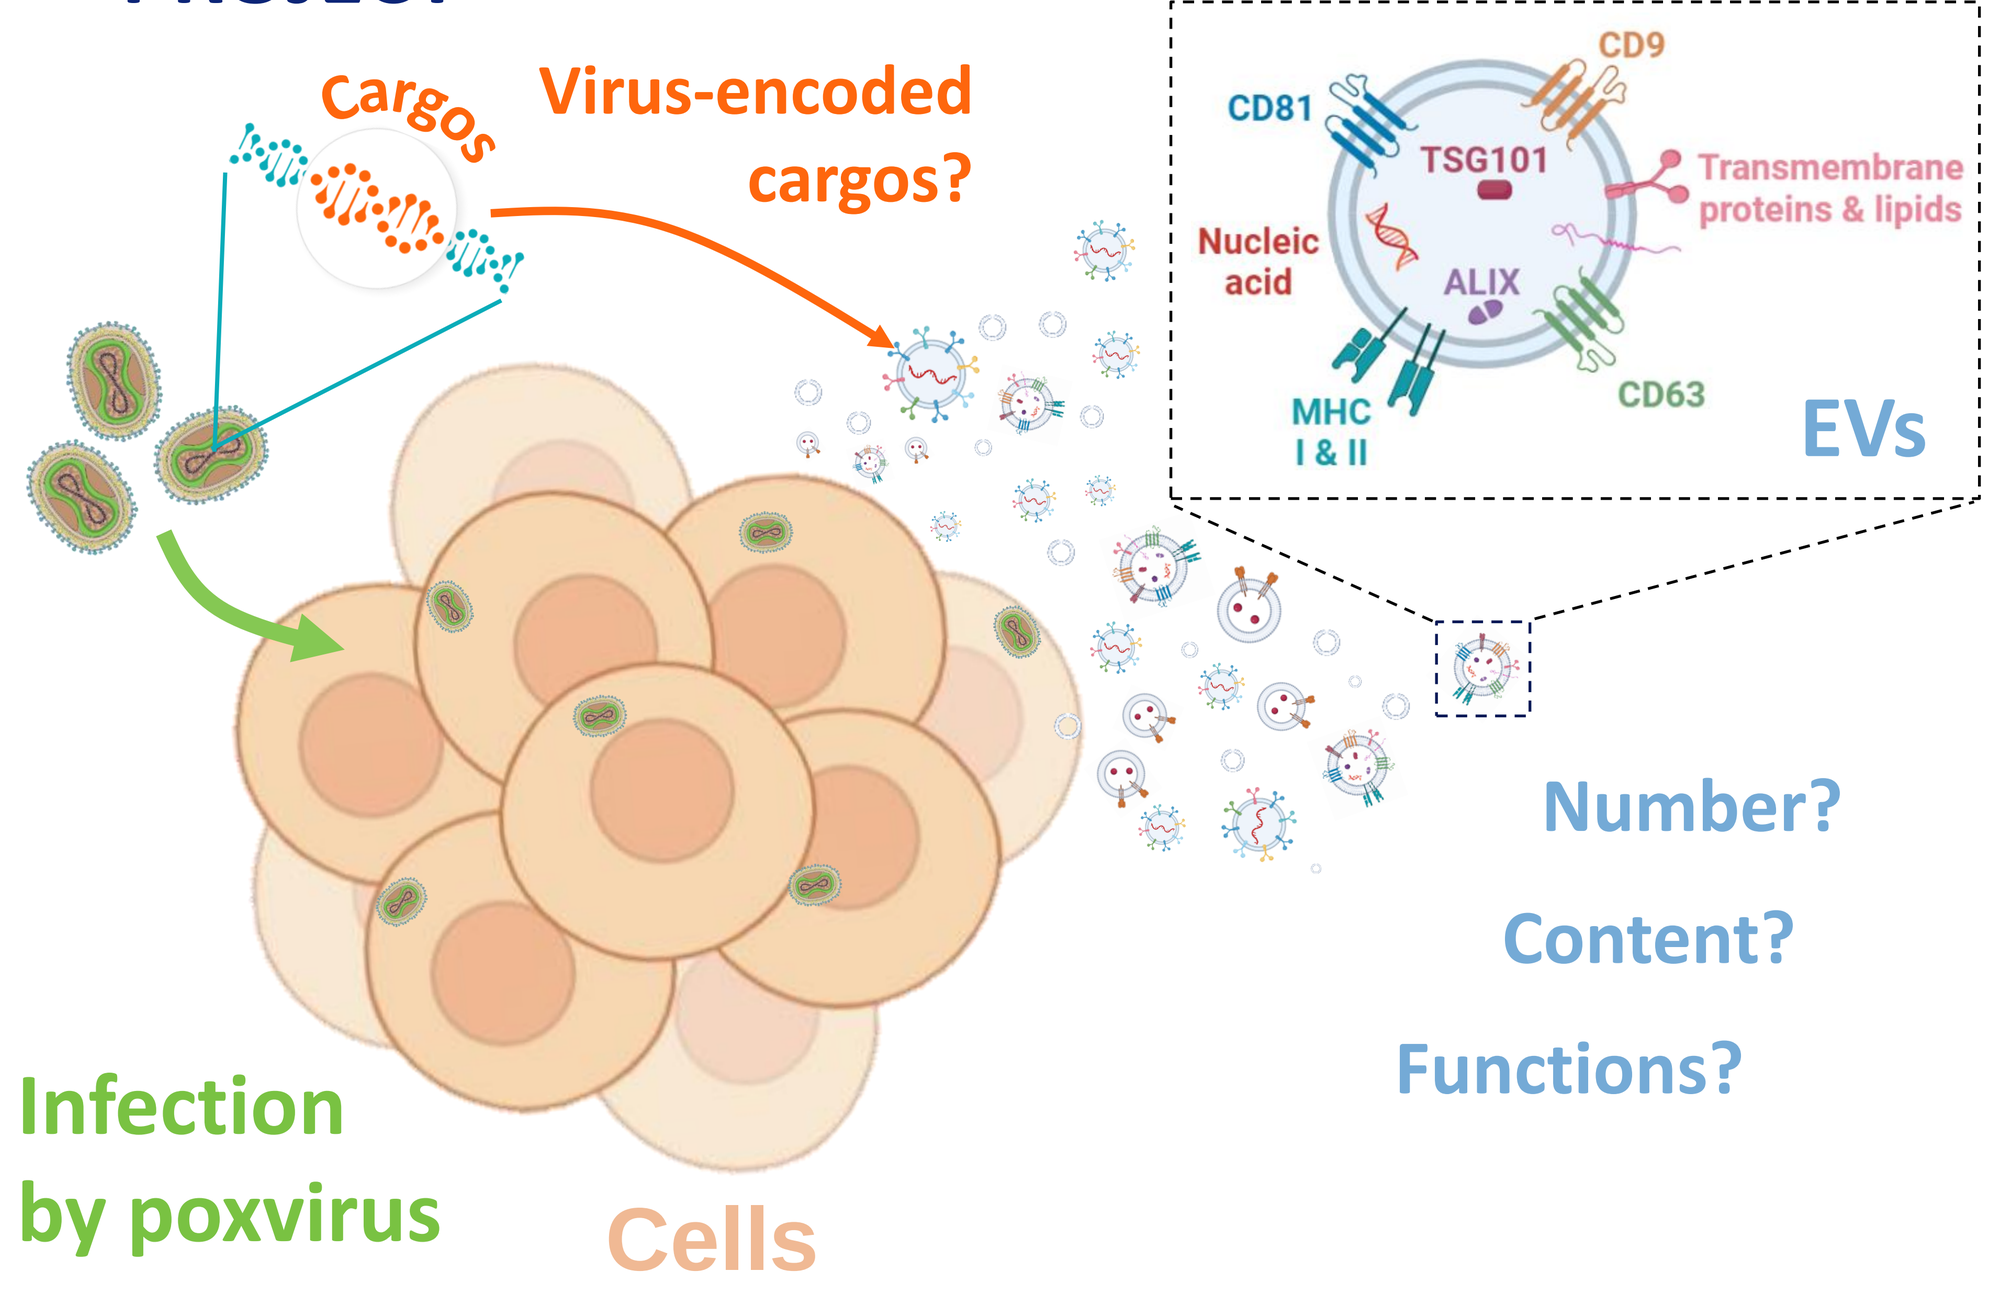

Cargos
Virus-encoded cargos?
CD81
CD9
TSG101
Transmembrane proteins & lipids
Nucleic acid
ALIX
MHC I & II
CD63
EVs
Number?
Content?
Functions?
Infection by poxvirus
Cells

MATERIAL & METHODS

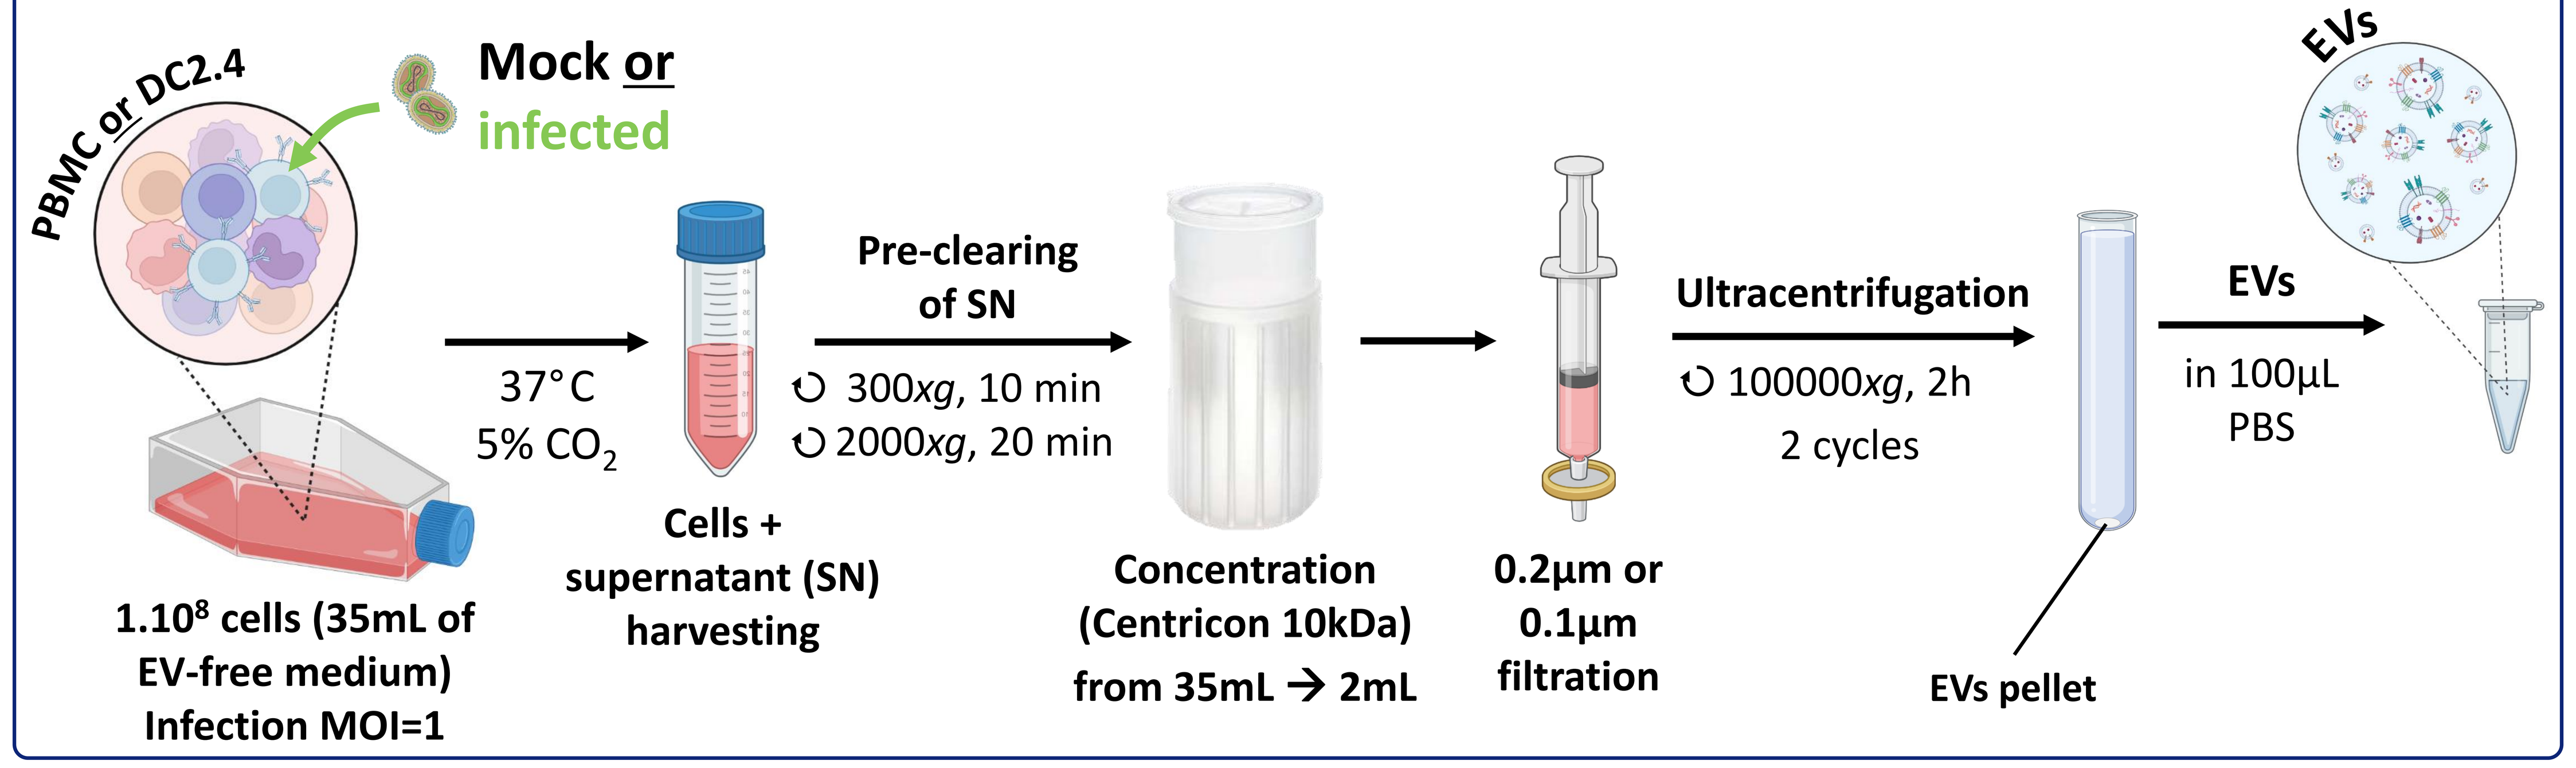

PBMC or DC2.4
Mock or infected
37°C
5% CO_2
1.10⁸ cells (35mL of EV-free medium) Infection MOI=1
Cells + supernatant (SN) harvesting
Pre-clearing of SN
300xg, 10 min
2000xg, 20 min
Concentration (Centricon 10kDa) from 35mL → 2mL
0.2µm or 0.1µm filtration
Ultracentrifugation
100000xg, 2h
2 cycles
EVs pellet
EVs
in 100µL PBS
EVs

RESULTS

❶ Isolation and separation of PBMC EVs from infectious poxviral particles

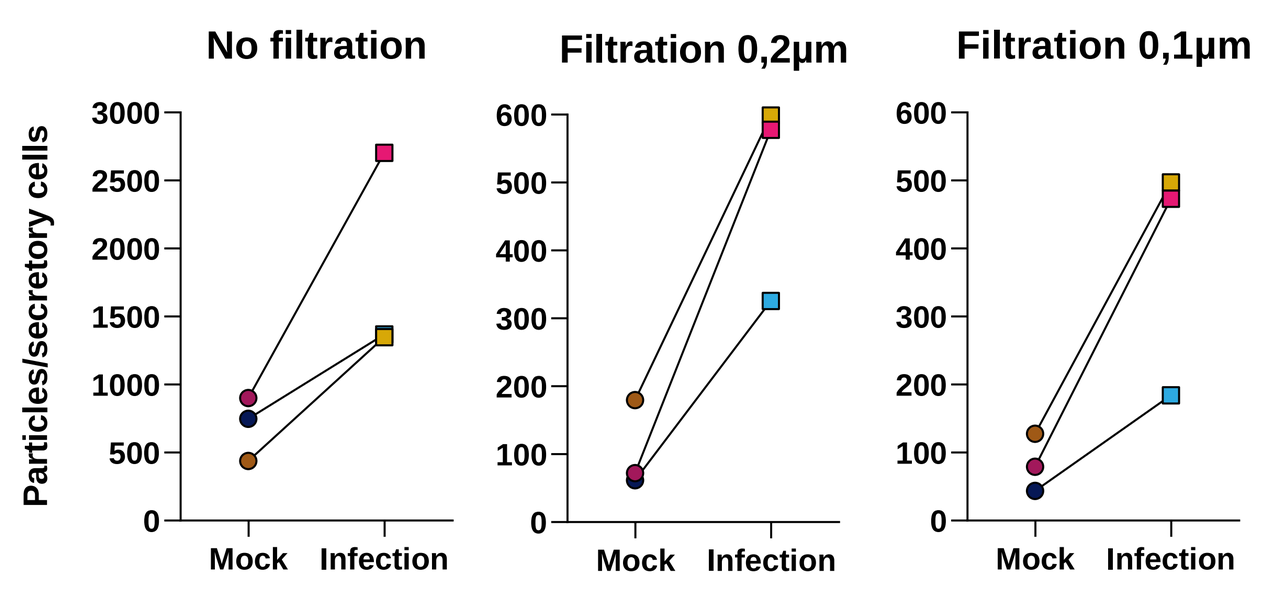

Concentration (EVs)
No filtration
Filtration 0,2µm
Filtration 0,1µm
Particles/secretory cells
Mock
Infection

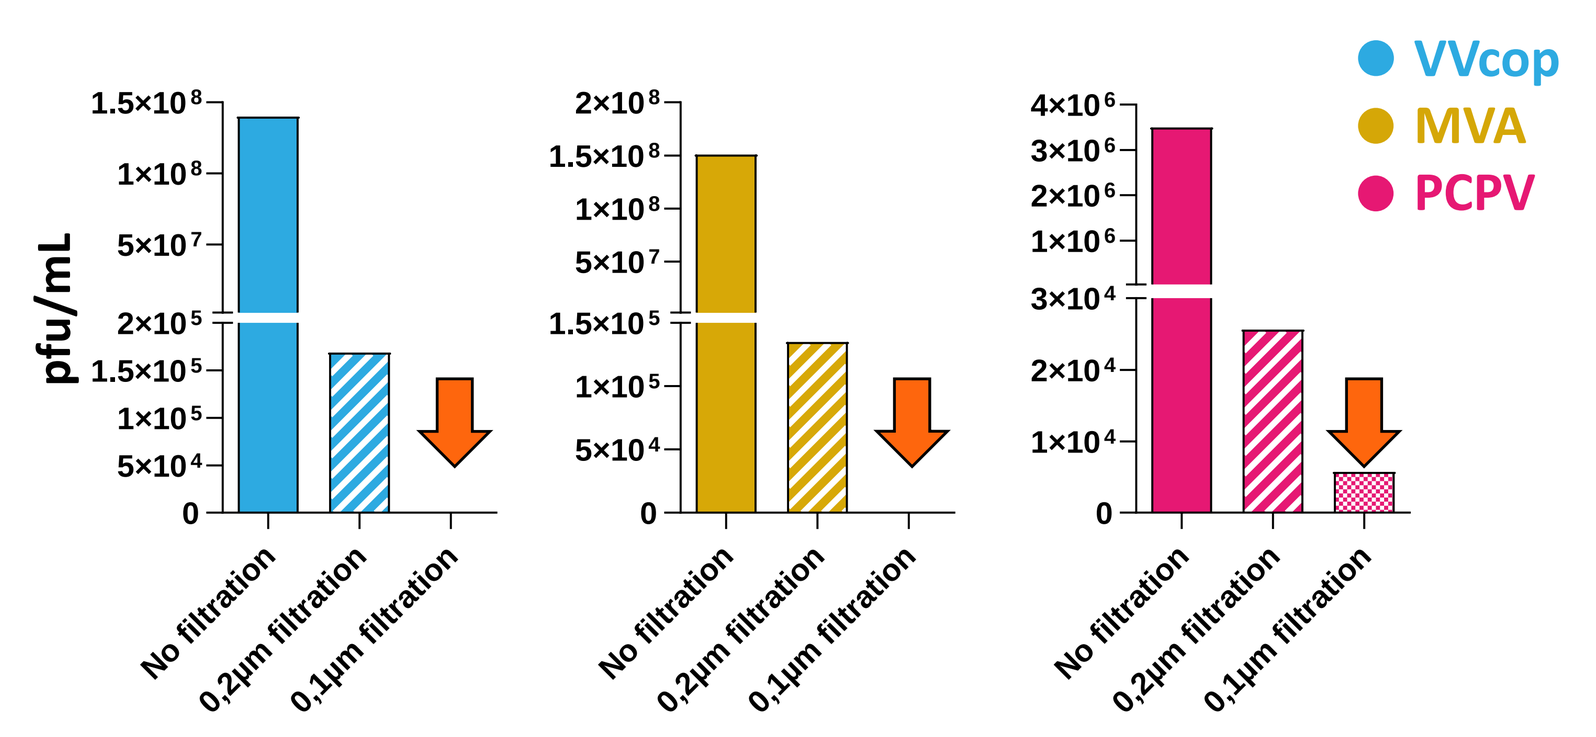

Titration (virus)
pfu/mL
No filtration
0,2µm filtration
0,1µm filtration
VVcop
MVA
PCPV

- The amount of EVs secreted by PBMC is increased after infection with all three viral strains tested
- Small EVs are separated from infectious viral particles by 0,1µm filtration (except for PCPV)

❷ Proteomic analysis of PBMC EVs by mass spectrometry

EVs isolated from PBMC mock or infected by MVA-GFP :
MOI = 1, +/- filtration, n = 4

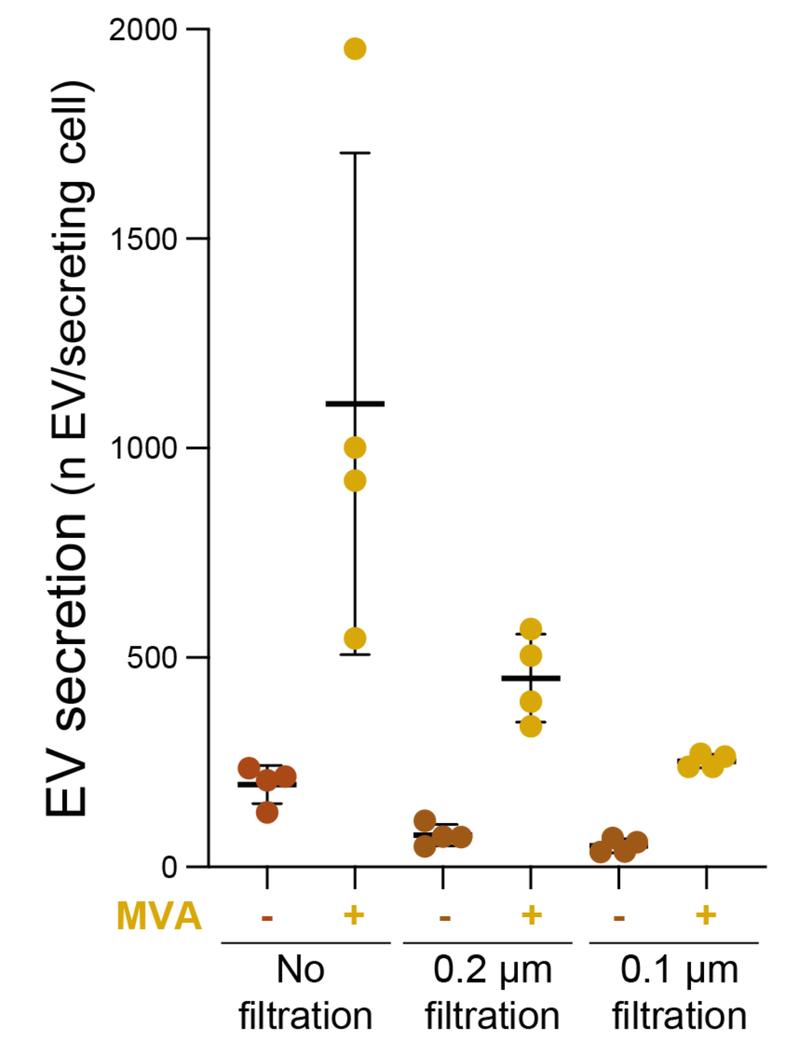

EV secretion (n EV/secreting cell)
MVA
No filtration
0.2 µm filtration
0.1 µm filtration

5x more EVs (+/- viral particles) in MVA-infected conditions

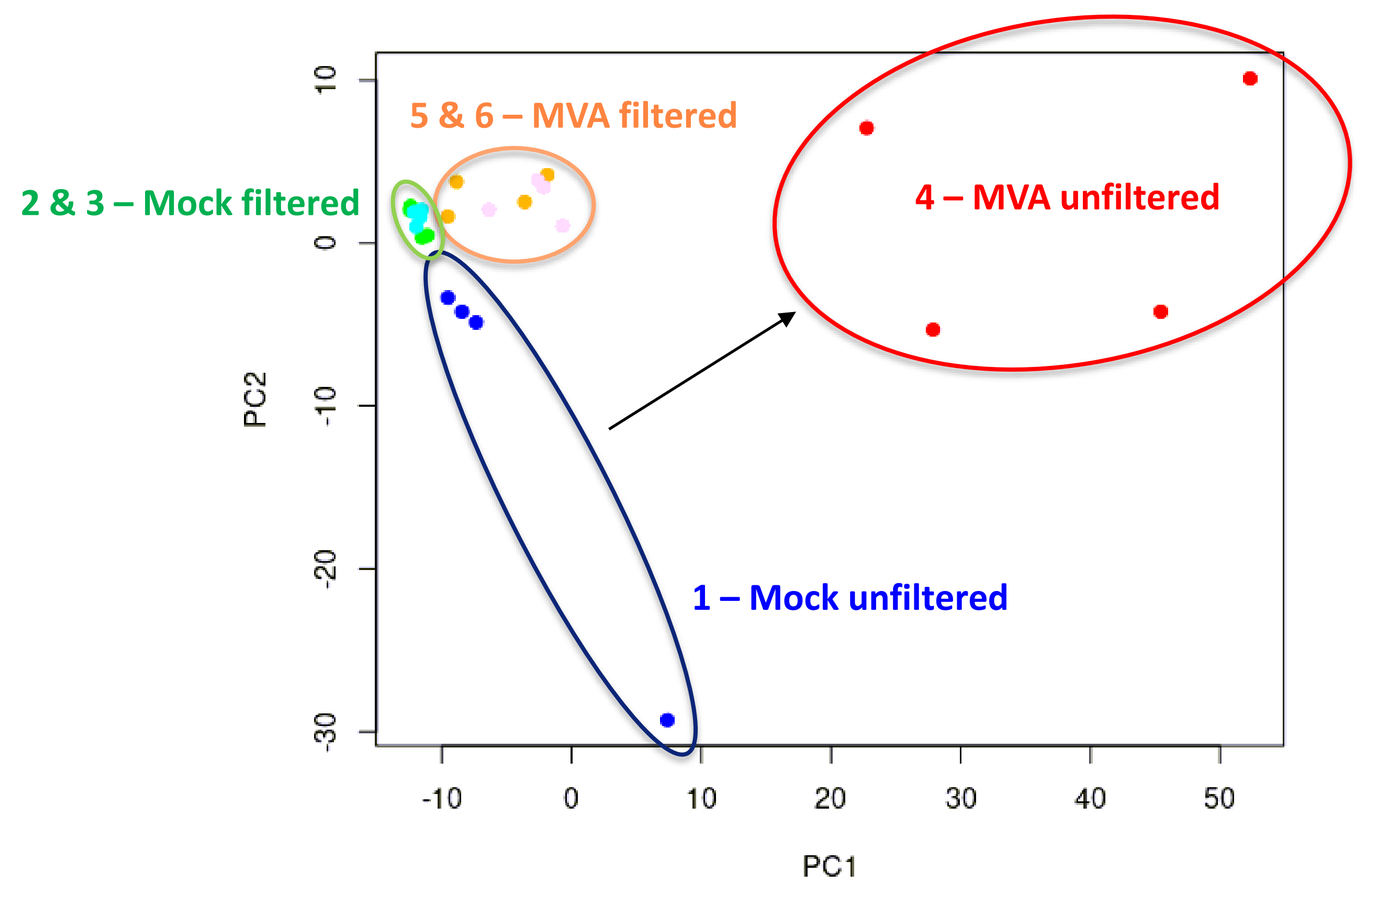

Principal Component Analysis
5 & 6 – MVA filtered
2 & 3 – Mock filtered
4 – MVA unfiltered
1 – Mock unfiltered
PC1
PC2

Effect of the infection on EVs protein abundance

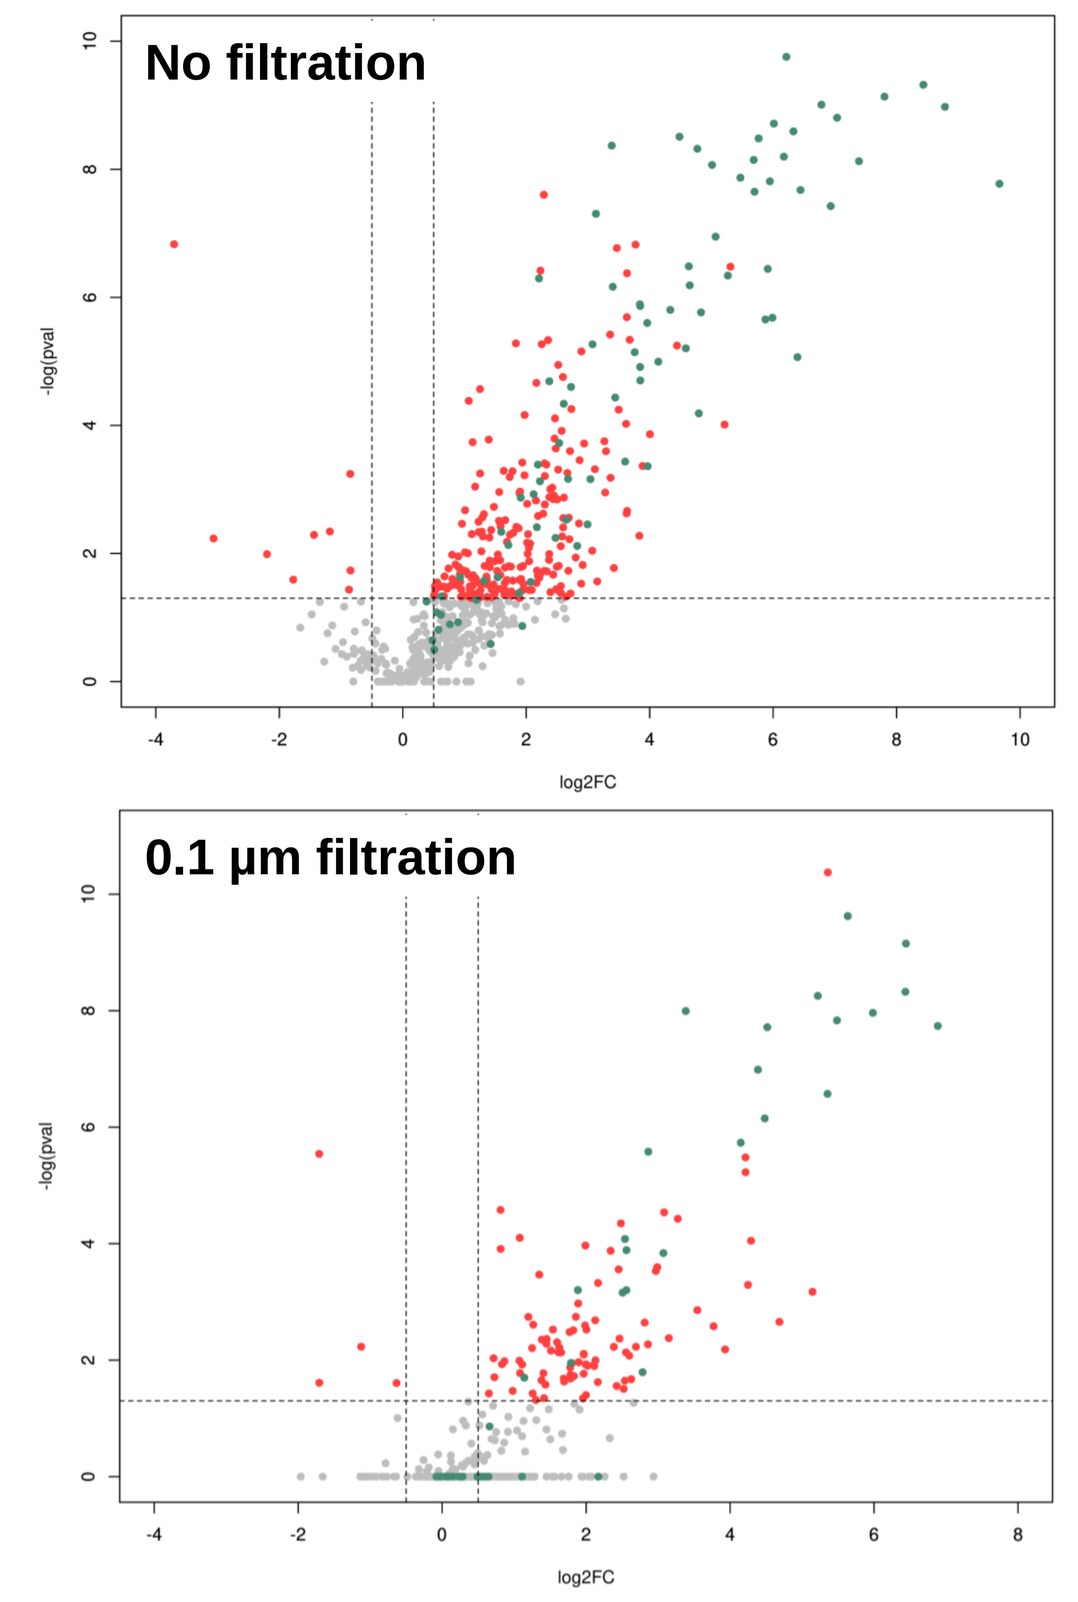

Changes in protein abundance
No filtration
0.1 µm filtration
log2FC
-log(pval

Differentially expressed viral and cellular proteins in EV fractions

❸ Isolation and characterization of 0.1µm-filtered EVs from murine dendritic DC2.4 cell line infected with MVA_pH5R-eGFP-OVA-fusion_p11k7.5_Cargo1_pH5R_Cargo2

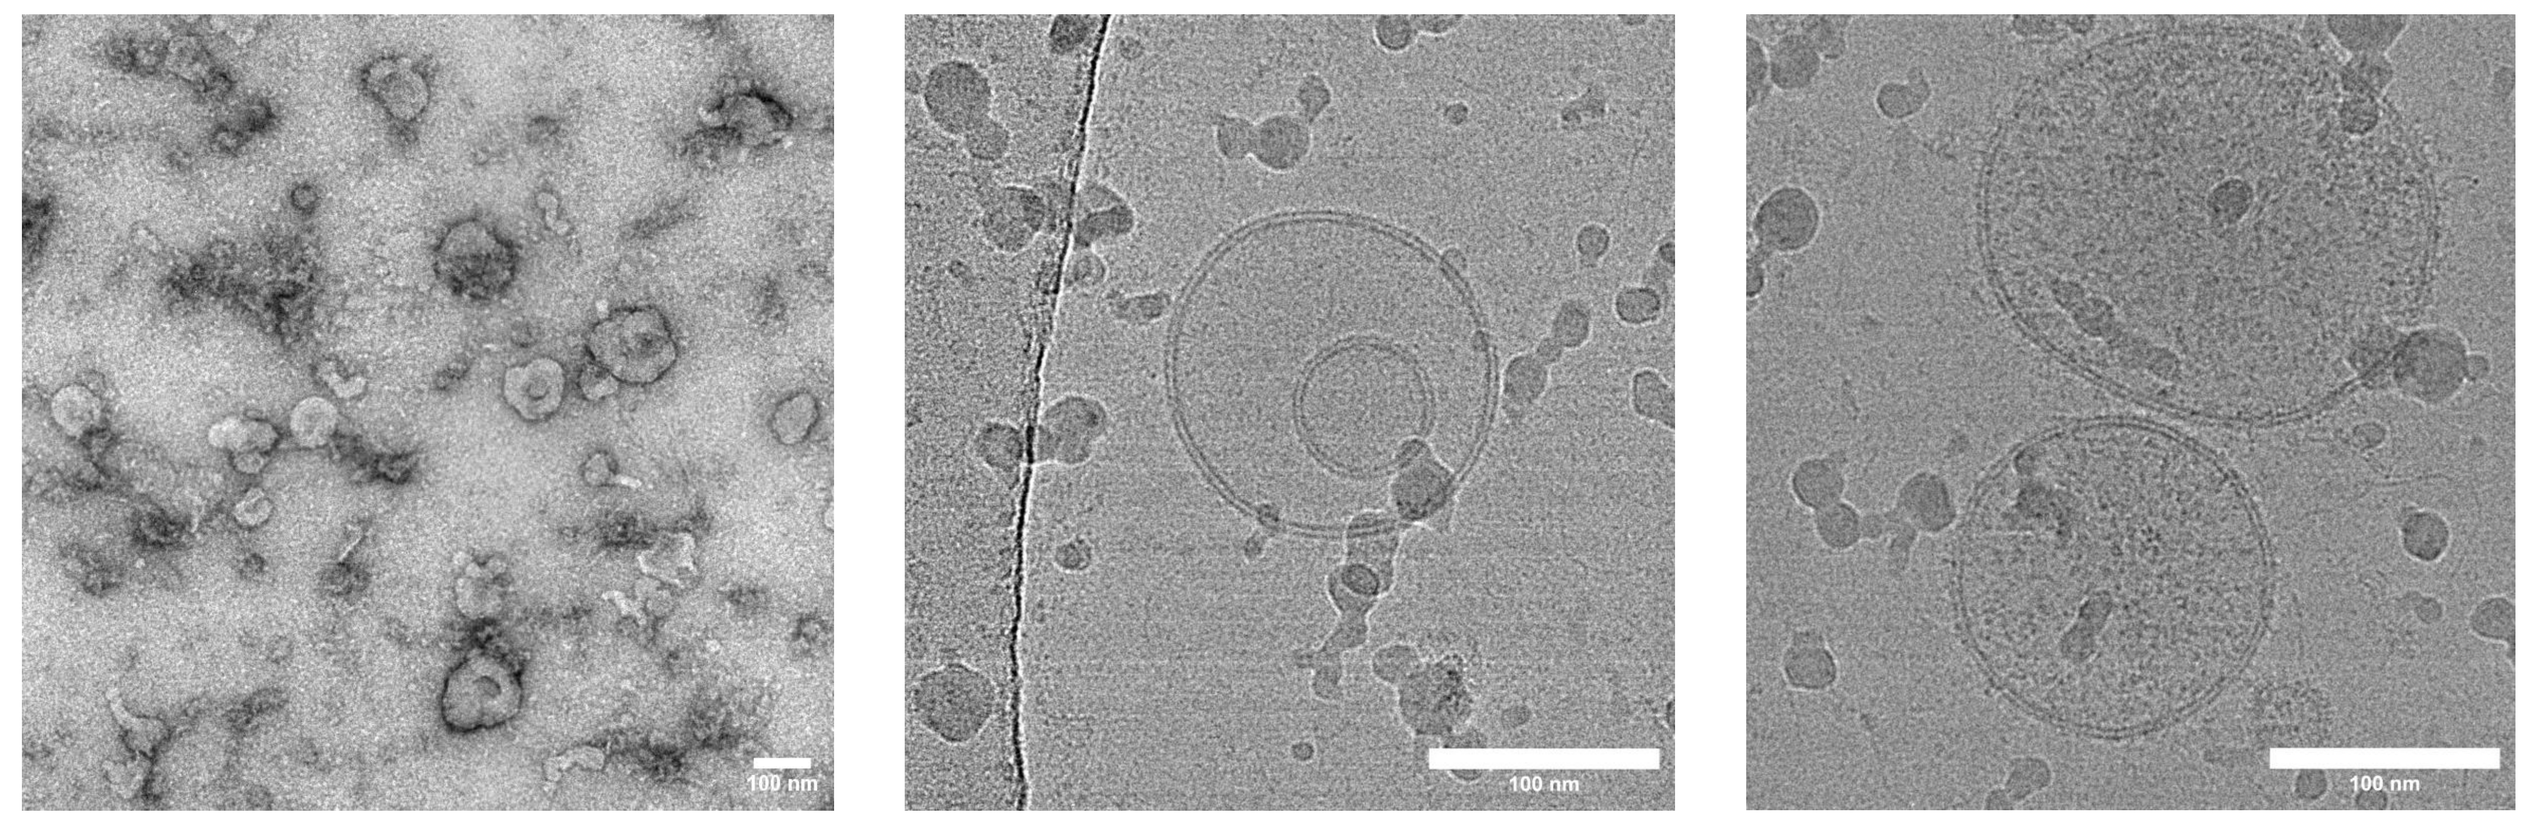

Classical cup-shaped vesicles in negative staining (TEM)

Bilayered vesicles in EV fraction were visualized (Cryo-EM)

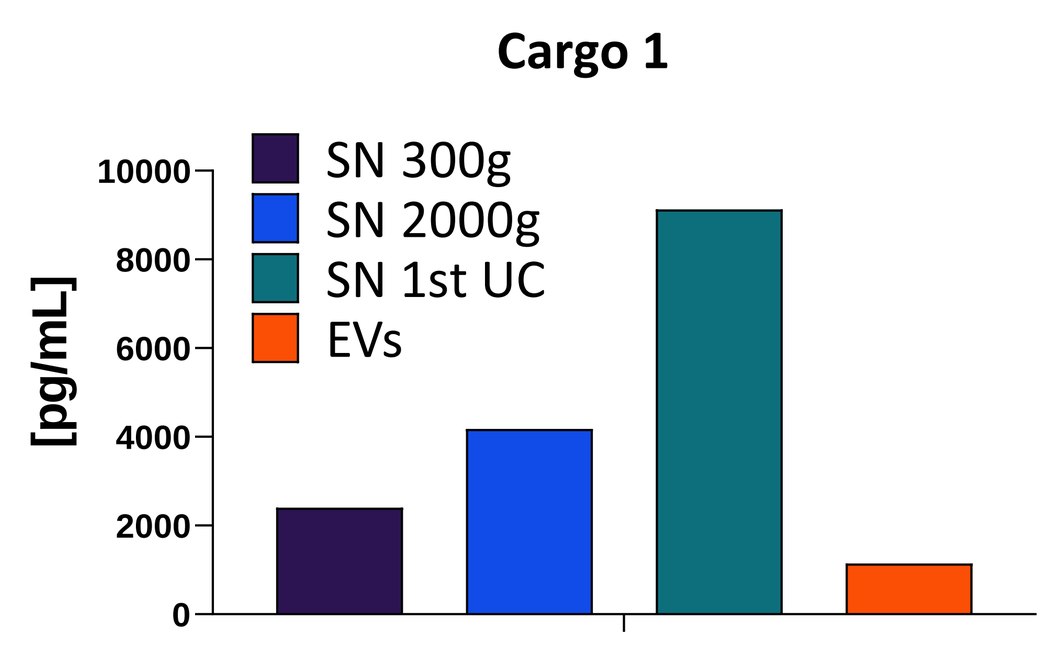

Cargo 1
[pg/mL]
SN 300g
SN 2000g
SN 1st UC
EVs

The soluble Cargo1 was detected in the EV fraction, suggesting it was associated to sedimentable structures

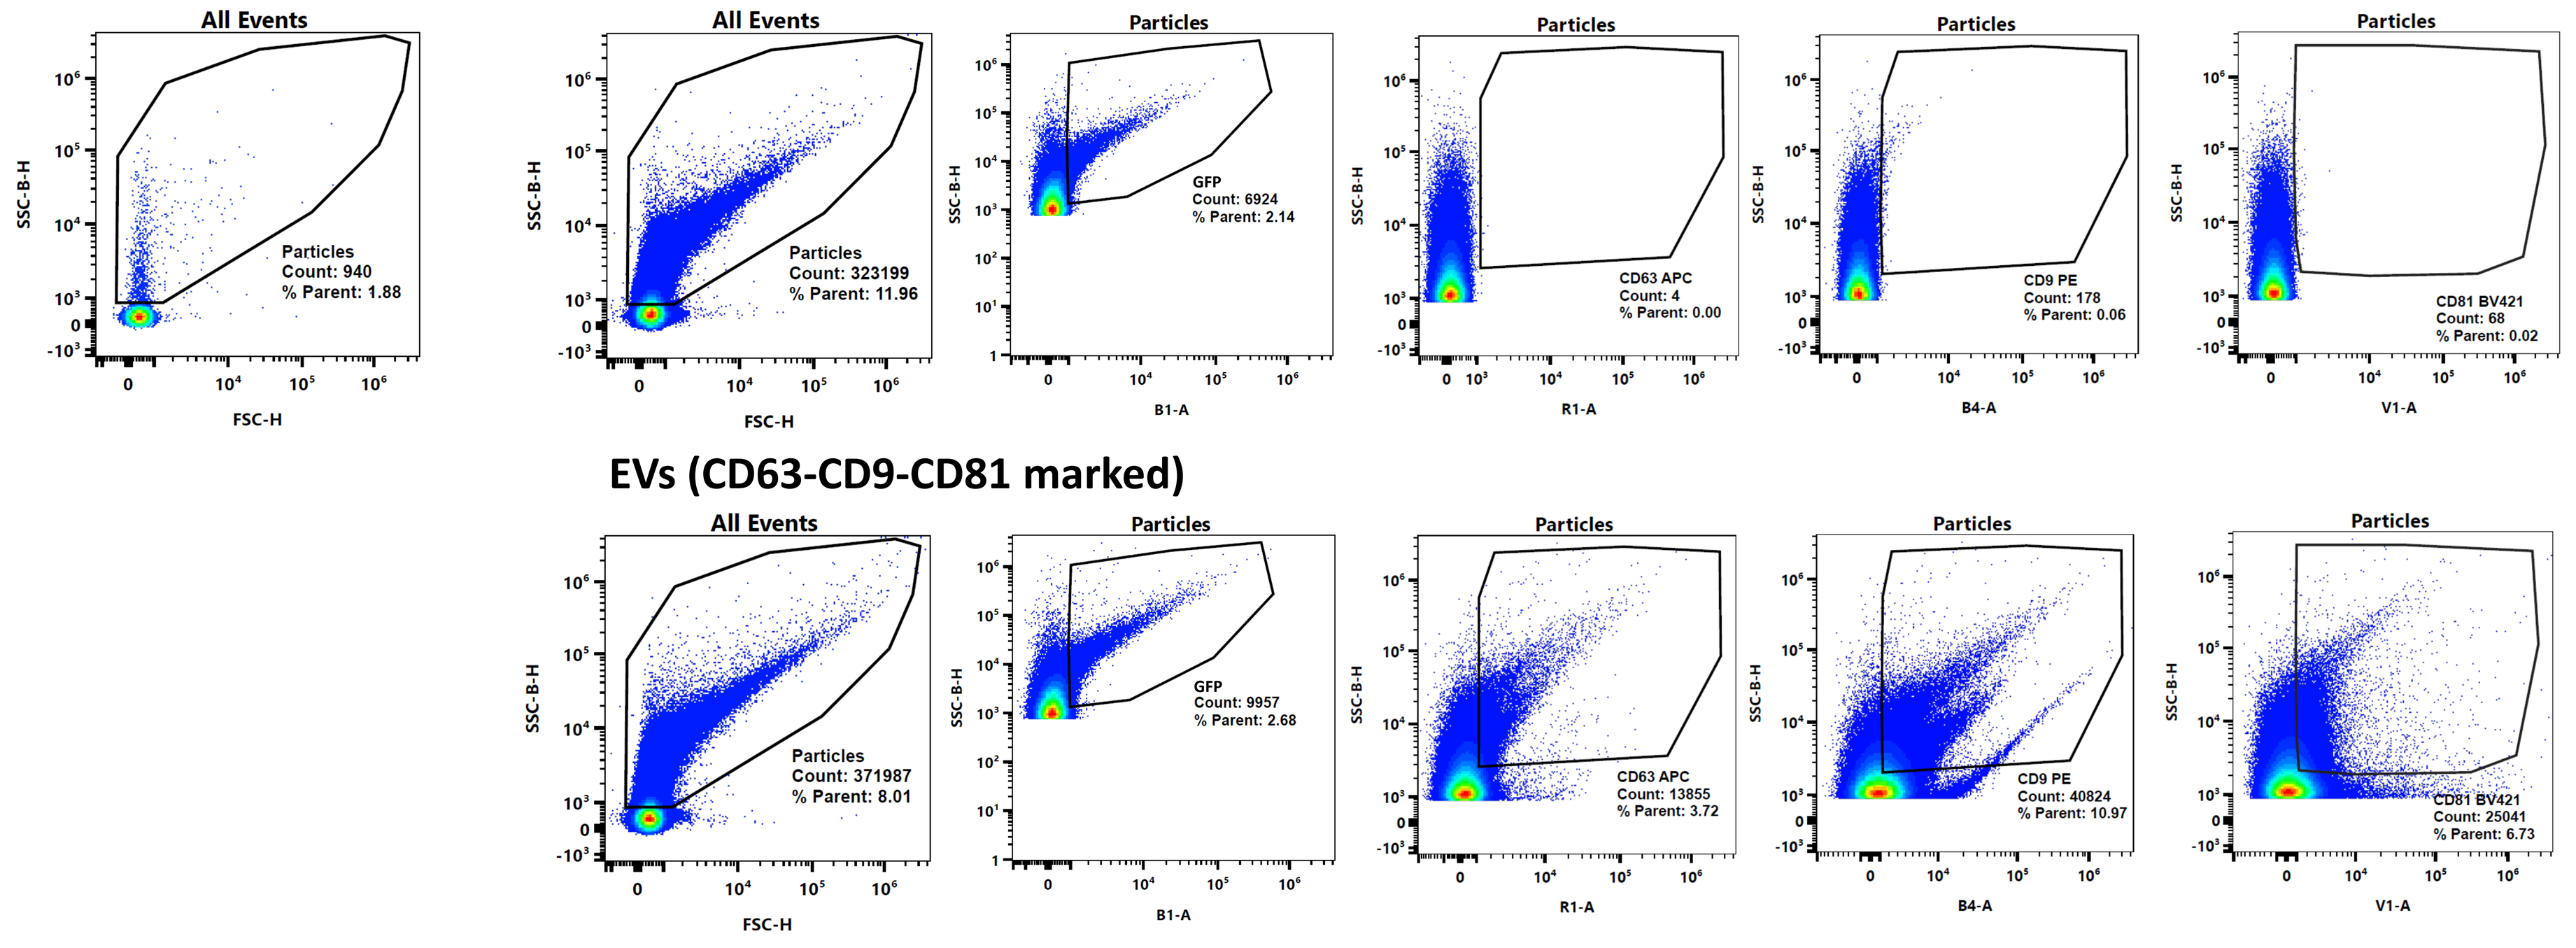

PBS 0.1µm filtered
EVs (non-marked)
EVs (CD63-CD9-CD81 marked)
All Events
Particles
GFP
CD63 APC
CD9 PE
CD81 BV421
SSC-B-H
FSC-H

First attempts of EV characterization by flow cytometry: EV preparations contain GFP-positive events. The tetraspanins CD63, CD9 and CD81 are detectable after staining with specific antibodies.

❹ Immunogenicity of isolated DC2.4 EVs (MVA-GFP-OVA-Cargo1-Cargo2) n=1

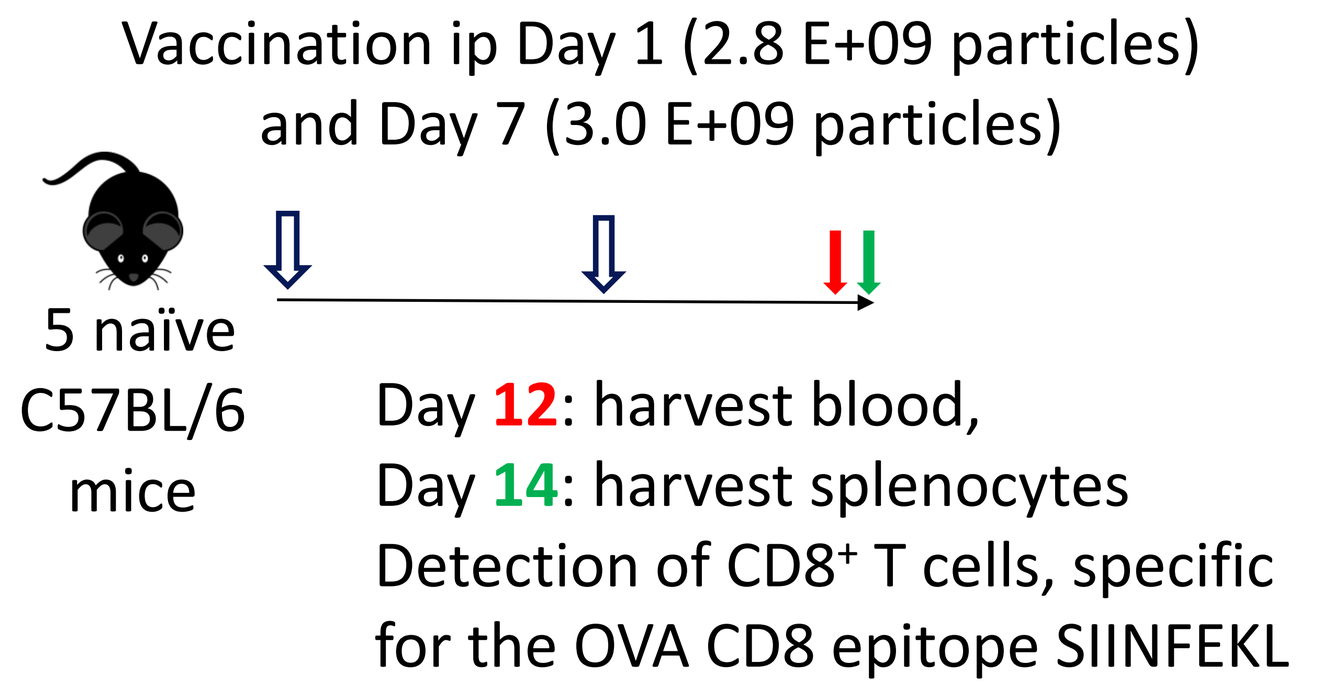

Vaccination ip Day 1 (2.8 E+09 particles) and Day 7 (3.0 E+09 particles)
5 naïve C57BL/6 mice
Day 12: harvest blood,
Day 14: harvest splenocytes
Detection of CD8+ T cells, specific for the OVA CD8 epitope SIINFEKL

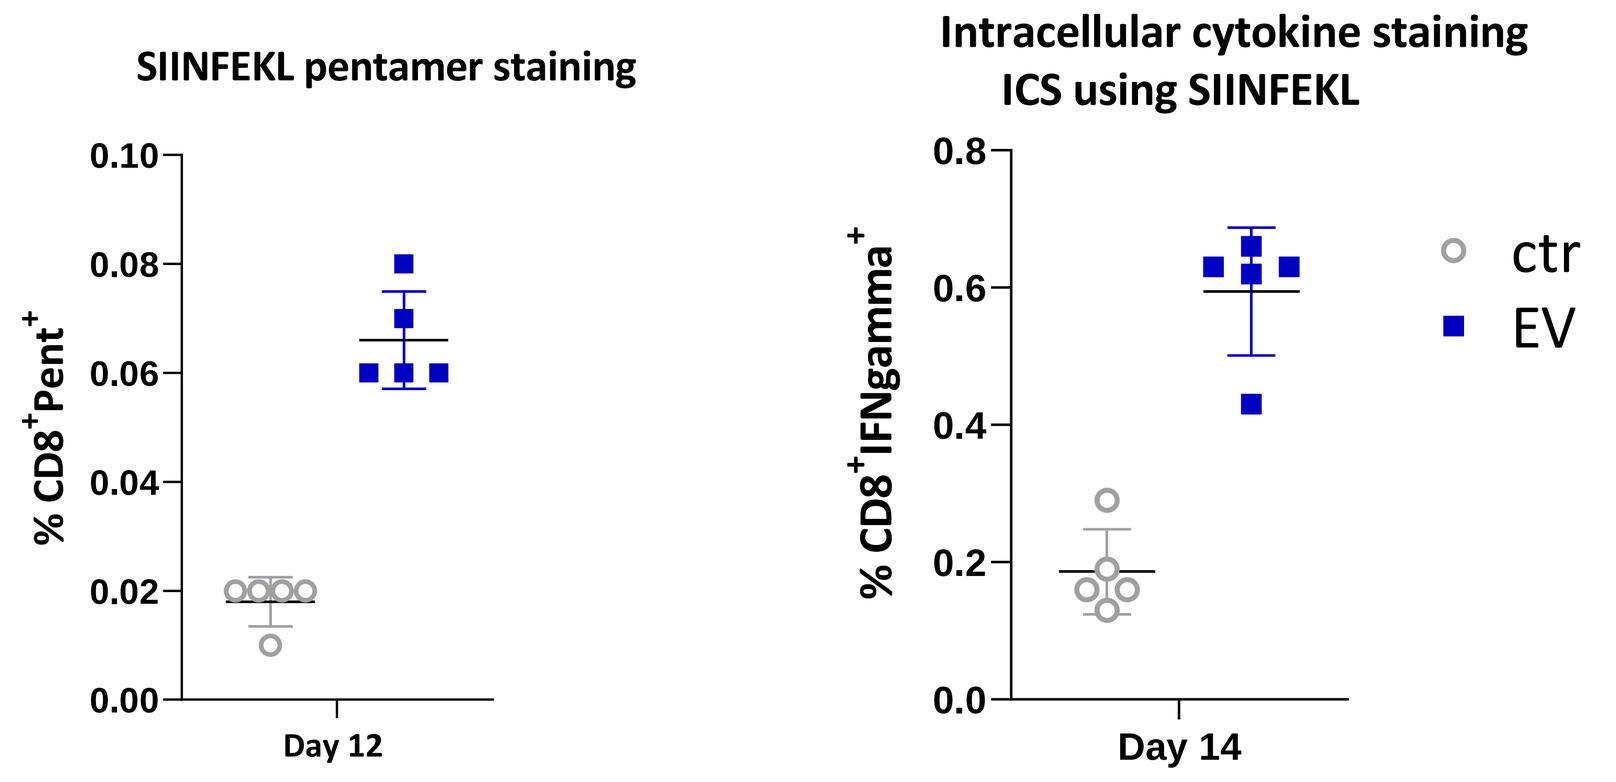

SIINFEKL pentamer staining
% CD8+Pent+
Day 12
Intracellular cytokine staining ICS using SIINFEKL
% CD8+IFNgamma+
Day 14
ctr
EV

Virus-free EVs from MVA-GFP-OVA-Cargo1-Cargo2 infected DC2.4 cells induce SIINFEKL-specific CD8+ T cells.

❺ Therapeutic efficacy of DC2.4 EVs (MVA-GFP-OVA-Cargo1-Cargo2) n=1

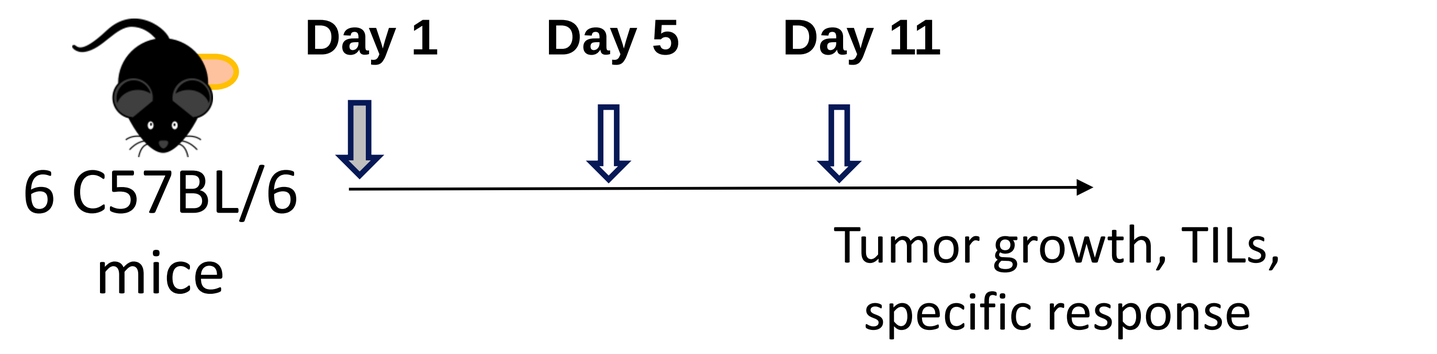

6 C57BL/6 mice
Day 1
Day 5
Day 11
Tumor growth, TILs, specific response

Day 1: E.G7-OVA (ATCC CRL-2113, a murine T lymphoblast cell line) cell grafting sc

Day 5 and **Day 11**: treatment with MVA (sc / sc) or EVs (sc/sc) or EVs (iv/iv).

MVA: MVA-GFP-OVA-Cargo1-Cargo2: 1x10^7 pfu.

EVs: virus-free EVs from DC2.4 infected with MVA-GFP-OVA-Cargo1-Cargo2.
Day 5: 4.0 E+09 particles. Day 11: 1.2 E+09 particles.

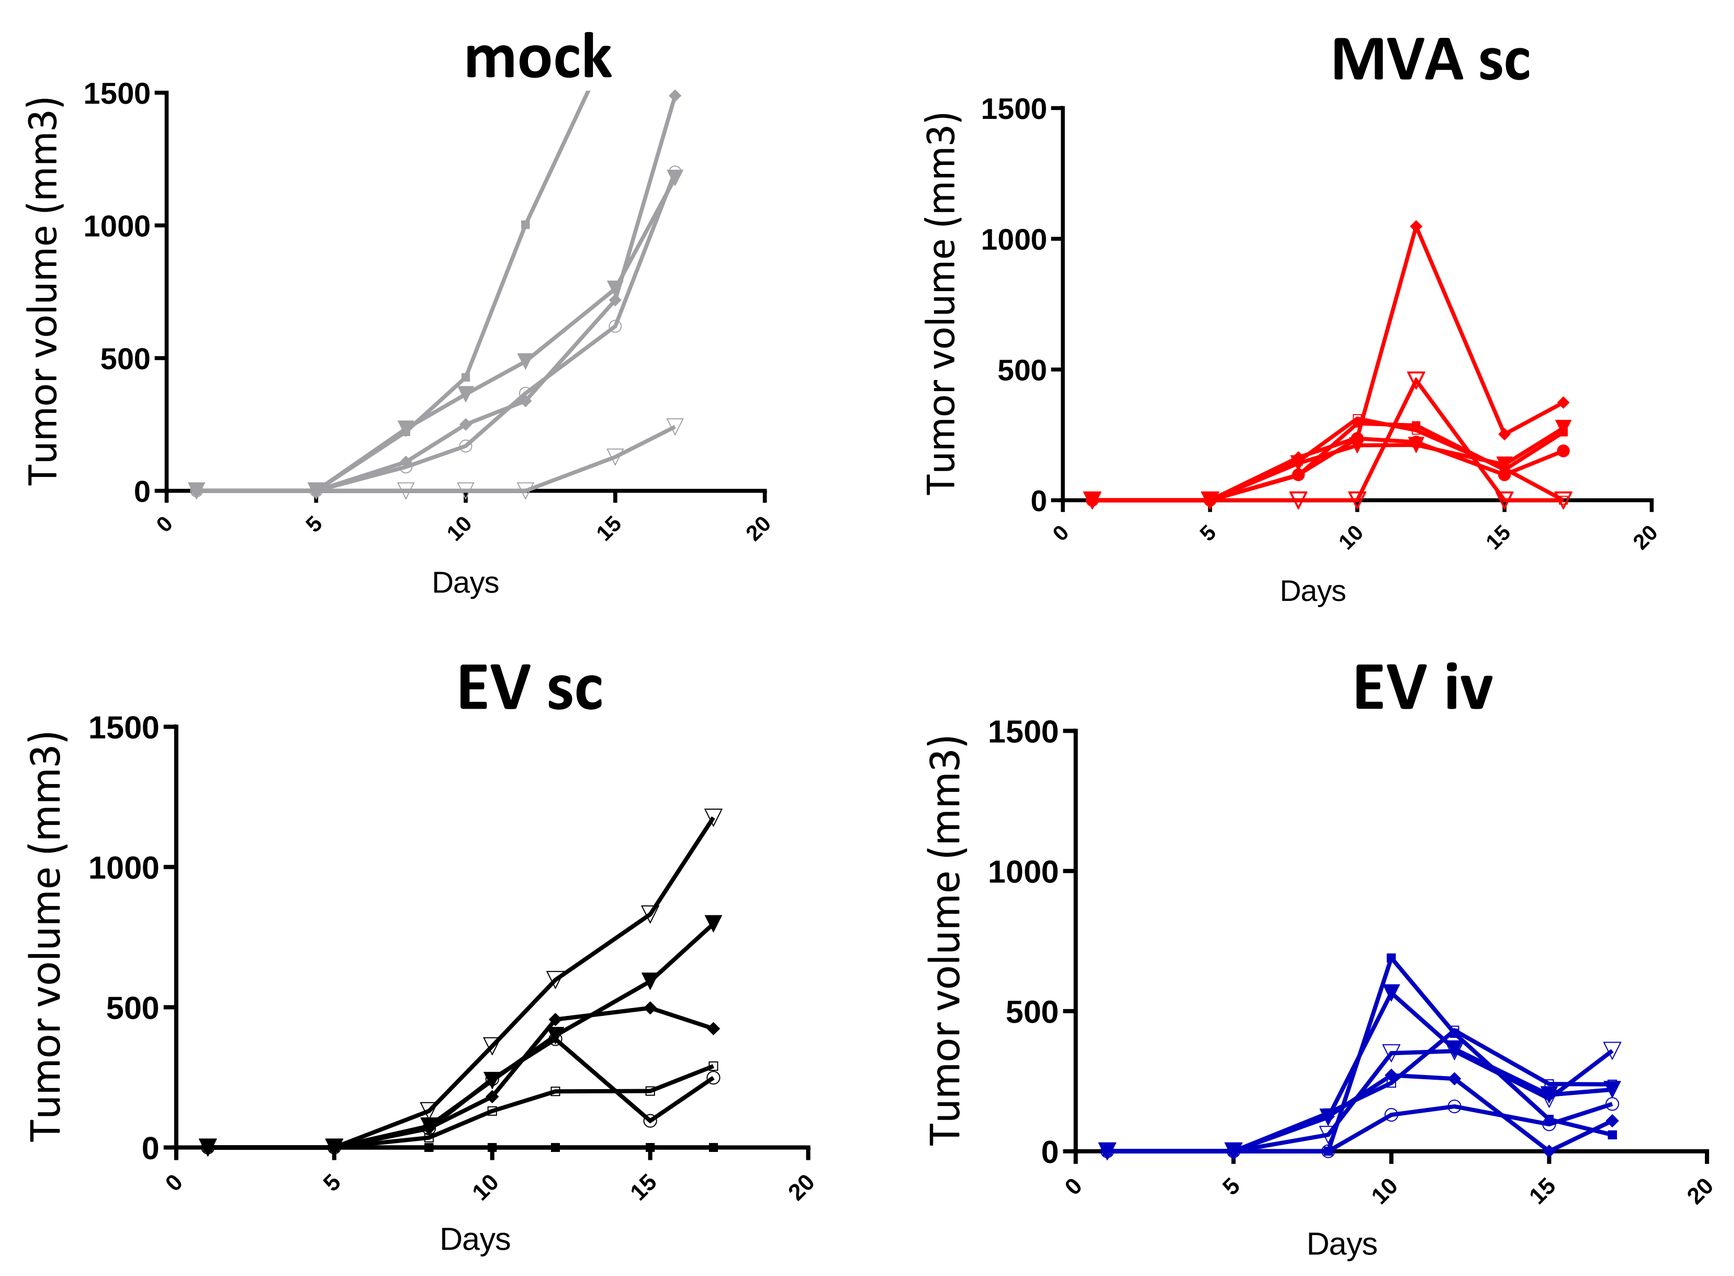

mock
MVA sc
EV sc
EV iv
Tumor volume (mm3)
Days

Virus-free EVs from MVA-GFP-OVA-Cargo 1-Cargo 2 infected DC2.4 showed therapeutic efficacy visible by a decrease in tumor burden after intravenous injection.

CONCLUSIONS

- **A protocol was established to separate small EVs from poxviruses.**
- **Poxviral infection increased EVs secretion from infected cells (PBMCs & DC2.4 cell line) and modified their protein content.**
- **Murine EVs isolated from DC2.4 cells, infected with an armed MVA-GFP-OVA virus, contain therapeutic cargo and GFP-OVA fusion protein.**
- **In vivo, such EVs generate OVA-specific CD8+ T cells in naïve mice and reduce tumor growth in EG.7-OVA tumor-bearing upon iv injection (n=1).**

PERSPECTIVES

- Avenue 1: **Ex vivo generation of loaded EVs in allogenic stem cells (off the shelf approach).**
- Avenue 2: **Design poxviral vectors which further enhance loaded EV secretion in vivo.**

DISCLOSURES

All authors affiliated to Transgene SA are or used to be employees of Transgene SA. Other authors do not have competing interest.